SITC 2024 | November 8-10; Houston, Texas

Abstract #283

Multiomic Profiling of LYL119: Reprogramming ROR1 CAR T cells With Reduced Exhaustion and Enhanced Memory Characteristics Is Associated With Increased AP-1 and Reduced NR4A Binding

Jia Lu, Viola C. Lam, Christina Cheung, Lora Zhao, Stefan Siebert, Ken Xiong, Sahithi Cheemalamarri, Christina Ta, Rowena Ellsworth, Purnima Sundar, Rachel C. Lynn, and Shobha Potluri

Lyell Immunopharma, Inc., South San Francisco, CA and Seattle, WA

For more information, please contact: jlu@lyell.com

Background

Effective solid tumor cell therapy requires new strategies to improve T-cell activation, persistence, and durable function[1-2]. Lyell has developed LYL119, a ROR1-targeted chimeric antigen receptor (CAR) T-cell product enhanced with four T-cell reprogramming technologies to improve CAR T-cell functionality[3].

Figure 1: LYL119, a ROR1-targeted CAR T-cell product

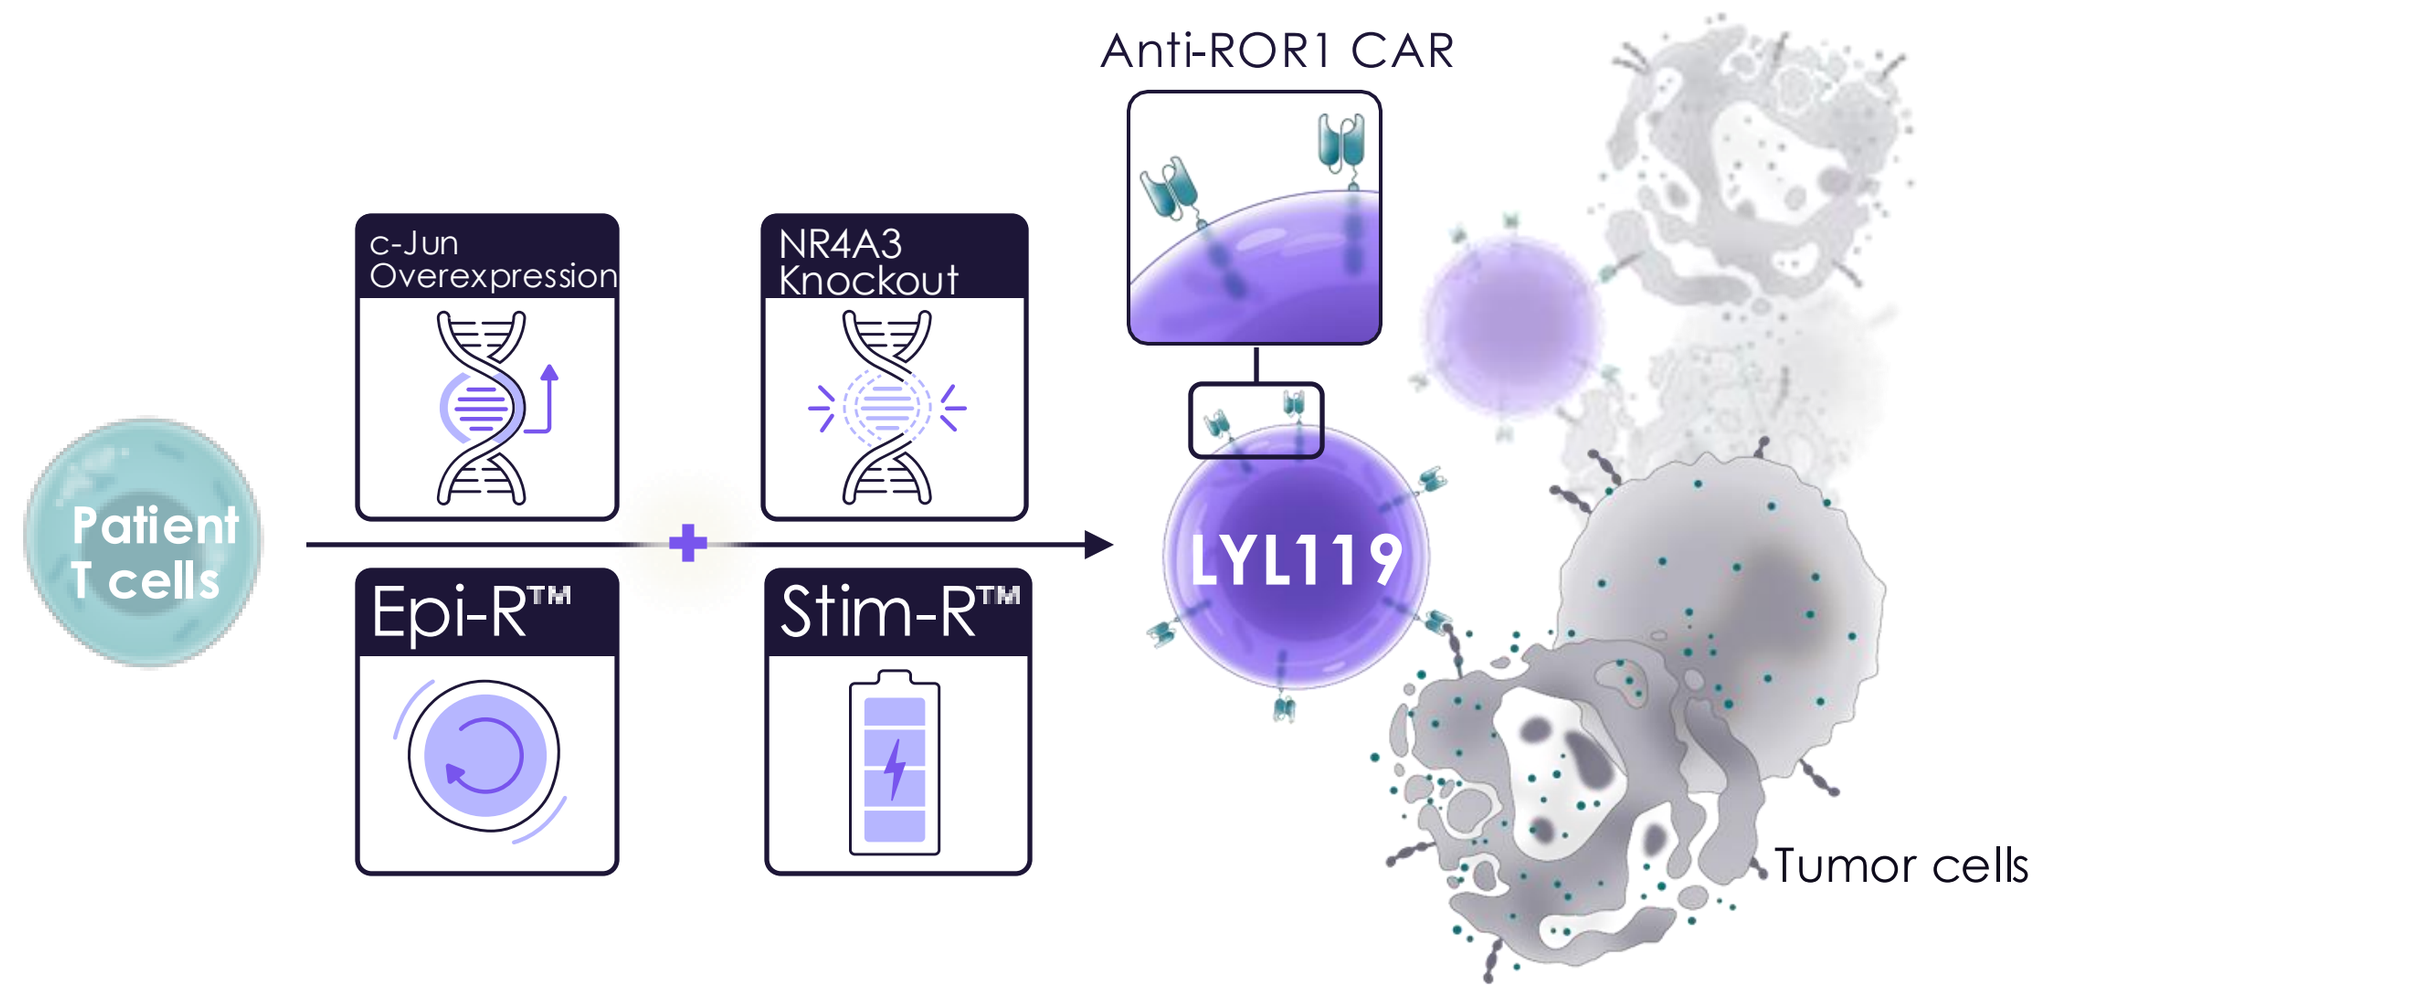

Anti-ROR1 CAR
c-Jun Overexpression
NR4A3 Knockout
Patient T cells
Epi-R™
Stim-R™
LYL119
Tumor cells

LYL119 is a ROR1-targeted CAR T-cell product incorporating (1) c-Jun overexpression to counter exhaustion; (2) NR4A3 knockout to further resist exhaustion and enhance antitumor activity; (3) Epi-R™ manufacturing protocol to preserve T-cell stem-like qualities; and (4) Stim-R™ activation technology (a synthetic biomimetic designed to precisely and physiologically present T-cell activation signals during manufacturing) to improve T-cell potency.

Methods

- LYL119 and control ROR1 CAR T cells (Non-edited + c-Jun + Epi-R) were manufactured with Epi-R and c-Jun overexpression. LYL119 was activated with Stim-R and incorporated NR4A3 editing. Control CAR T cells were activated with a standard reagent and were not edited.
- ROR1 CAR T-cell functional activity as well as transcriptional and epigenetic profiles (single-cell Multiome) were evaluated in vitro after a serial restimulation assay designed to induce exhaustion.
- Antitumor activity and transcriptional profile (CITE-Seq) of ROR1 CAR T cells were evaluated in vivo using a ROR1-expressing H1975 human NSCLC xenograft model in NSG MHCI/II dKO mice.

Results

LYL119 demonstrates superior in vitro and in vivo activity compared to control (Figures 2 and 3)

Figure 2: LYL119 demonstrates superior in vitro activity following serial antigen restimulation

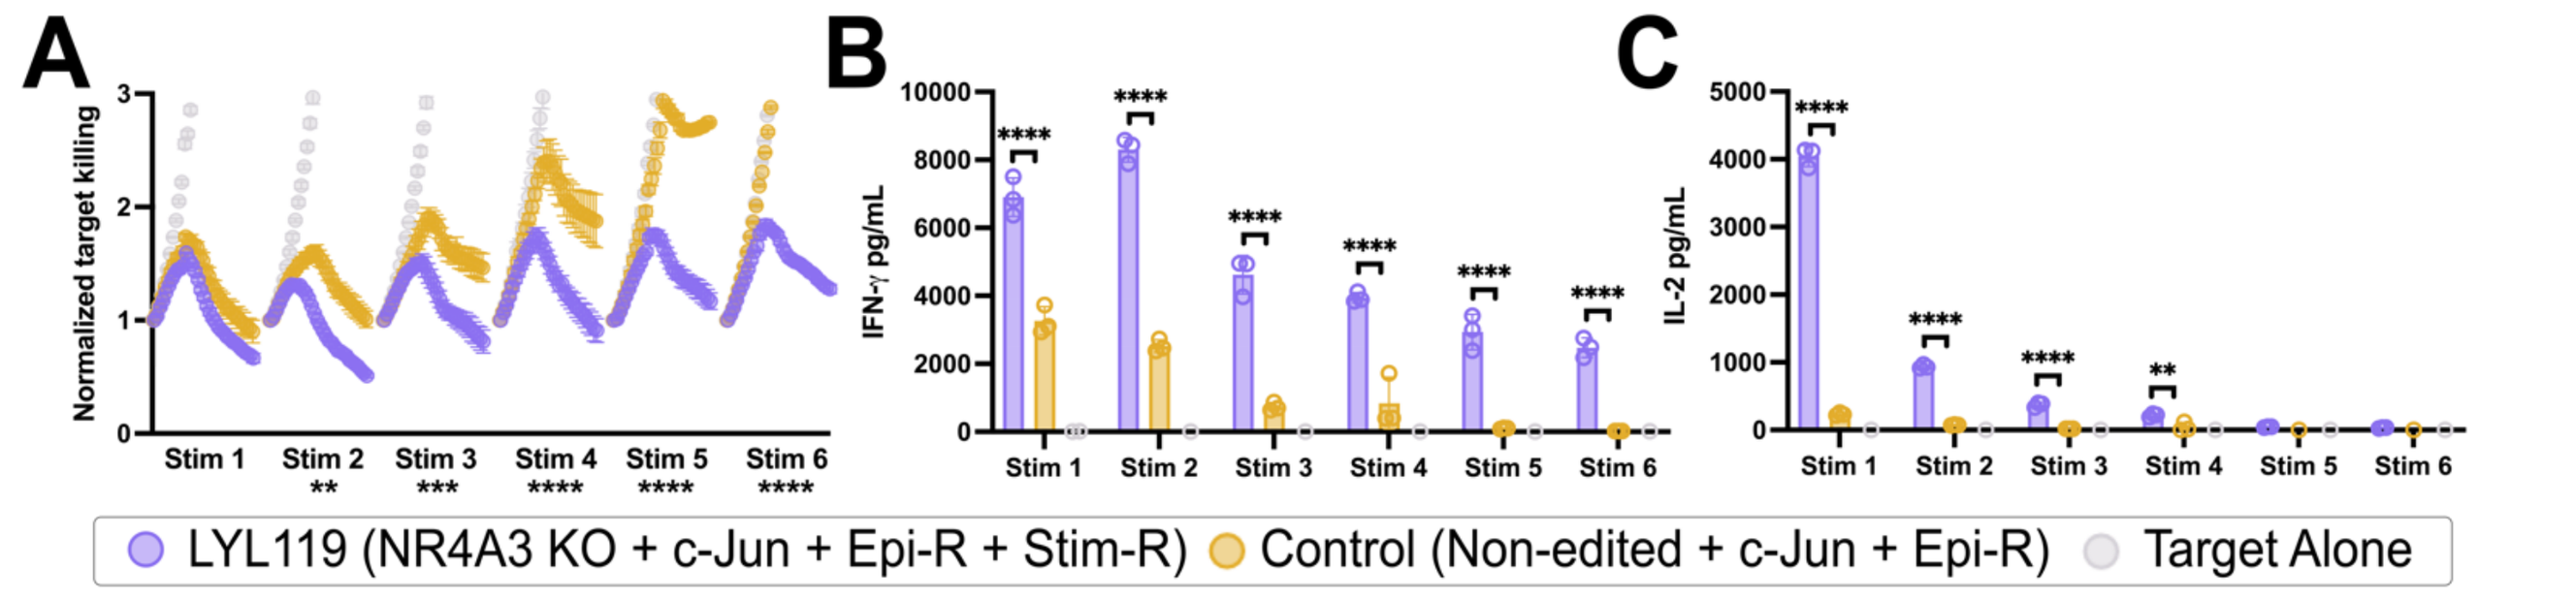

A
Normalized target killing
Stim 1
Stim 2 **
Stim 3 ***
Stim 4 ****
Stim 5 ****
Stim 6 ****
B
IFN-γ pg/mL
C
IL-2 pg/mL
LYL119 (NR4A3 KO + c-Jun + Epi-R + Stim-R)
Control (Non-edited + c-Jun + Epi-R)
Target Alone

Serial antigen restimulation of ROR1 CAR T cells with A549 NSCLC tumor cells at an E:T ratio of 1:4. (A) Cytotoxicity, (B) IFN-γ and (C) IL-2 secretion from one out of two representative products is shown. Error bars represent mean ± SD of triplicate wells. Asterisks indicate significant differences comparing (A) the AUC, (B) IFN-γ, or (C) IL-2 levels of LYL119 and control CAR T cells.

Figure 3: LYL119 eliminates xenograft tumor and prolongs animal survival in vivo at low CAR T-cell dose

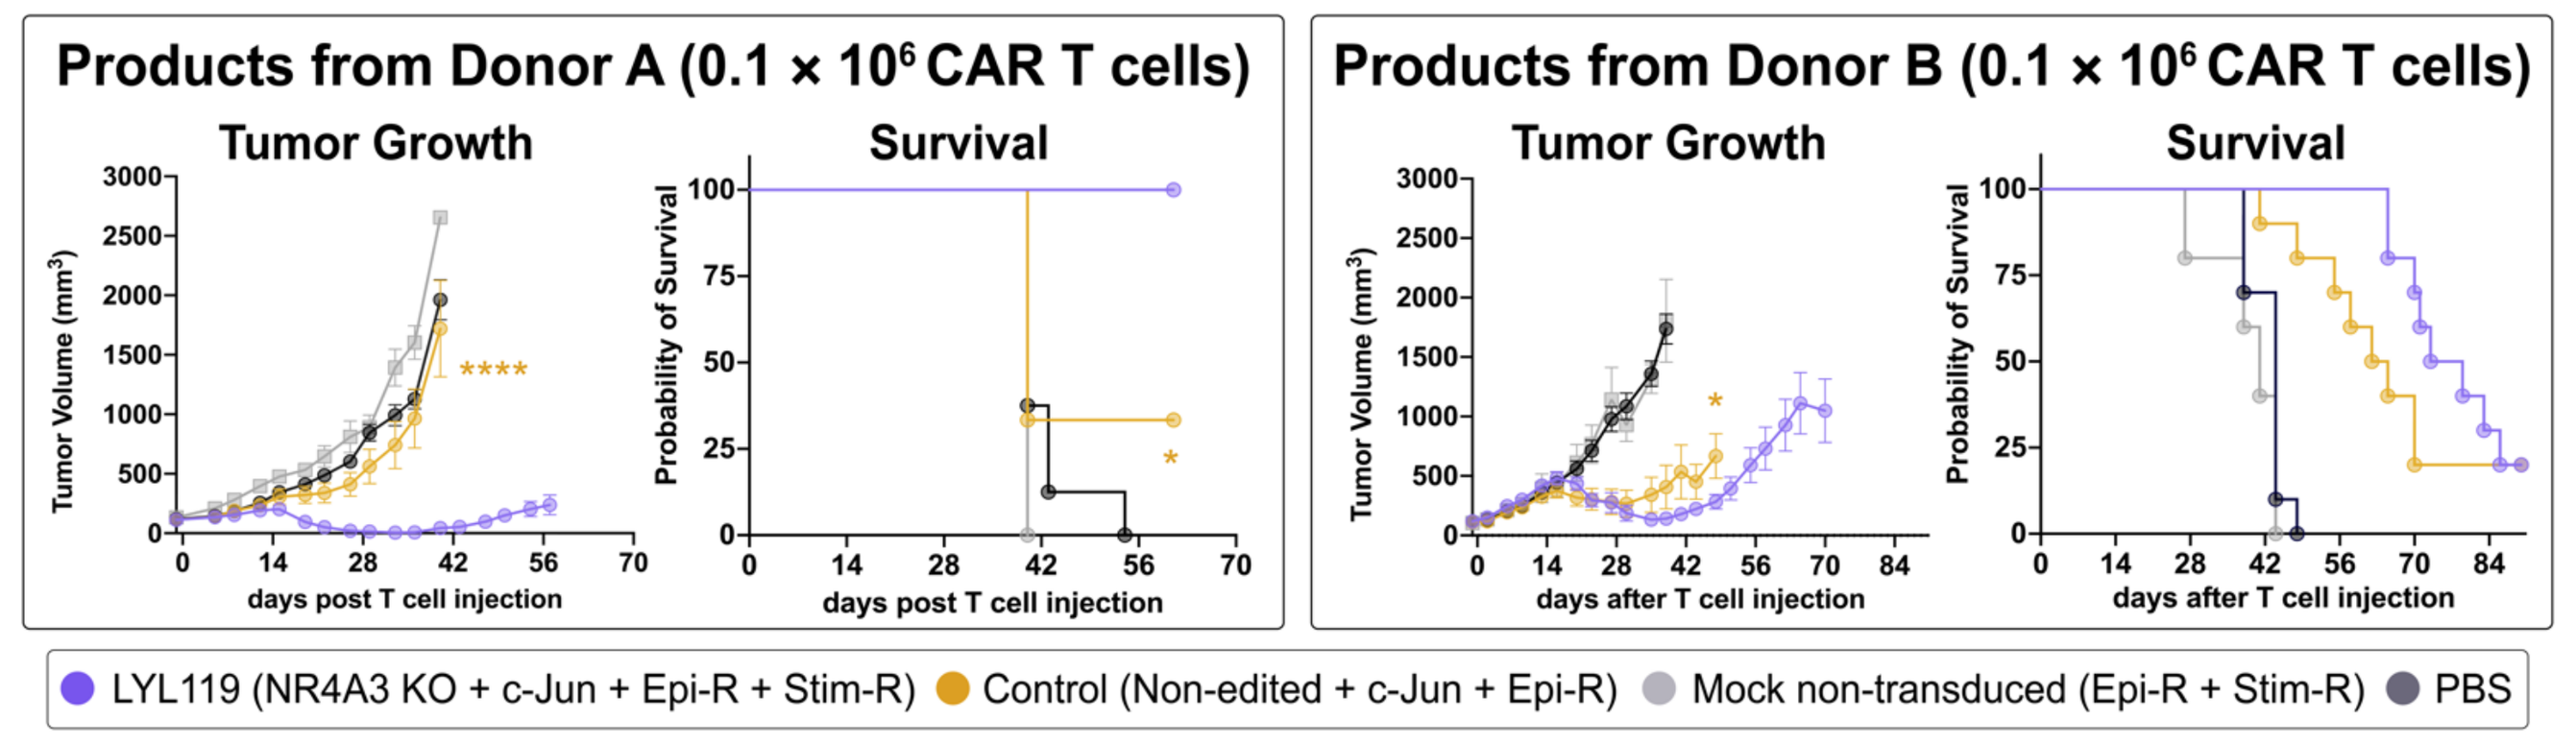

Products from Donor A (0.1 × 10^6 CAR T cells)
Tumor Growth
Survival
Tumor Volume (mm³)
Probability of Survival
days post T cell injection
Products from Donor B (0.1 × 10^6 CAR T cells)
days after T cell injection
LYL119 (NR4A3 KO + c-Jun + Epi-R + Stim-R)
Control (Non-edited + c-Jun + Epi-R)
Mock non-transduced (Epi-R + Stim-R)
PBS

Tumor volume and animal survival at the 0.1 x 10^6 CAR T-cell dose in a H1975 xenograft NSG MHCI/II dKO mouse model. Data from 2 of 3 independent animal studies are shown. Error bars represent mean ± SEM. Asterisks indicate significant differences compared to LYL119-treated animals.

LYL119 exhibits an improved phenotype associated with increased AP-1 motif accessibility and reduced NR4A motif accessibility following in vitro antigen restimulation (Figure 4 to 6)

After 15 days of restimulation with A549 NSCLC tumor cells, compared to Control, LYL119 displays:

- Lower proportion of terminally exhausted T cells and downregulation of exhaustion-related gene signatures across phenotypic cell subsets, associated with reduced NR4A motif accessibility (Figure 4 to 5)
- Upregulation of memory T-cell marker IL7R across phenotypic cell subsets, associated with increased AP-1 motif accessibility (Figure 6, A to D)
- Presence of a unique TCF7-hi stem-like cell subset with upregulation of stemness-related and effector-related gene signatures, indicating both persistence and cytotoxicity (Figure 6E)

Figure 4: LYL119 exhibits reduced exhaustion-related gene signatures in vitro

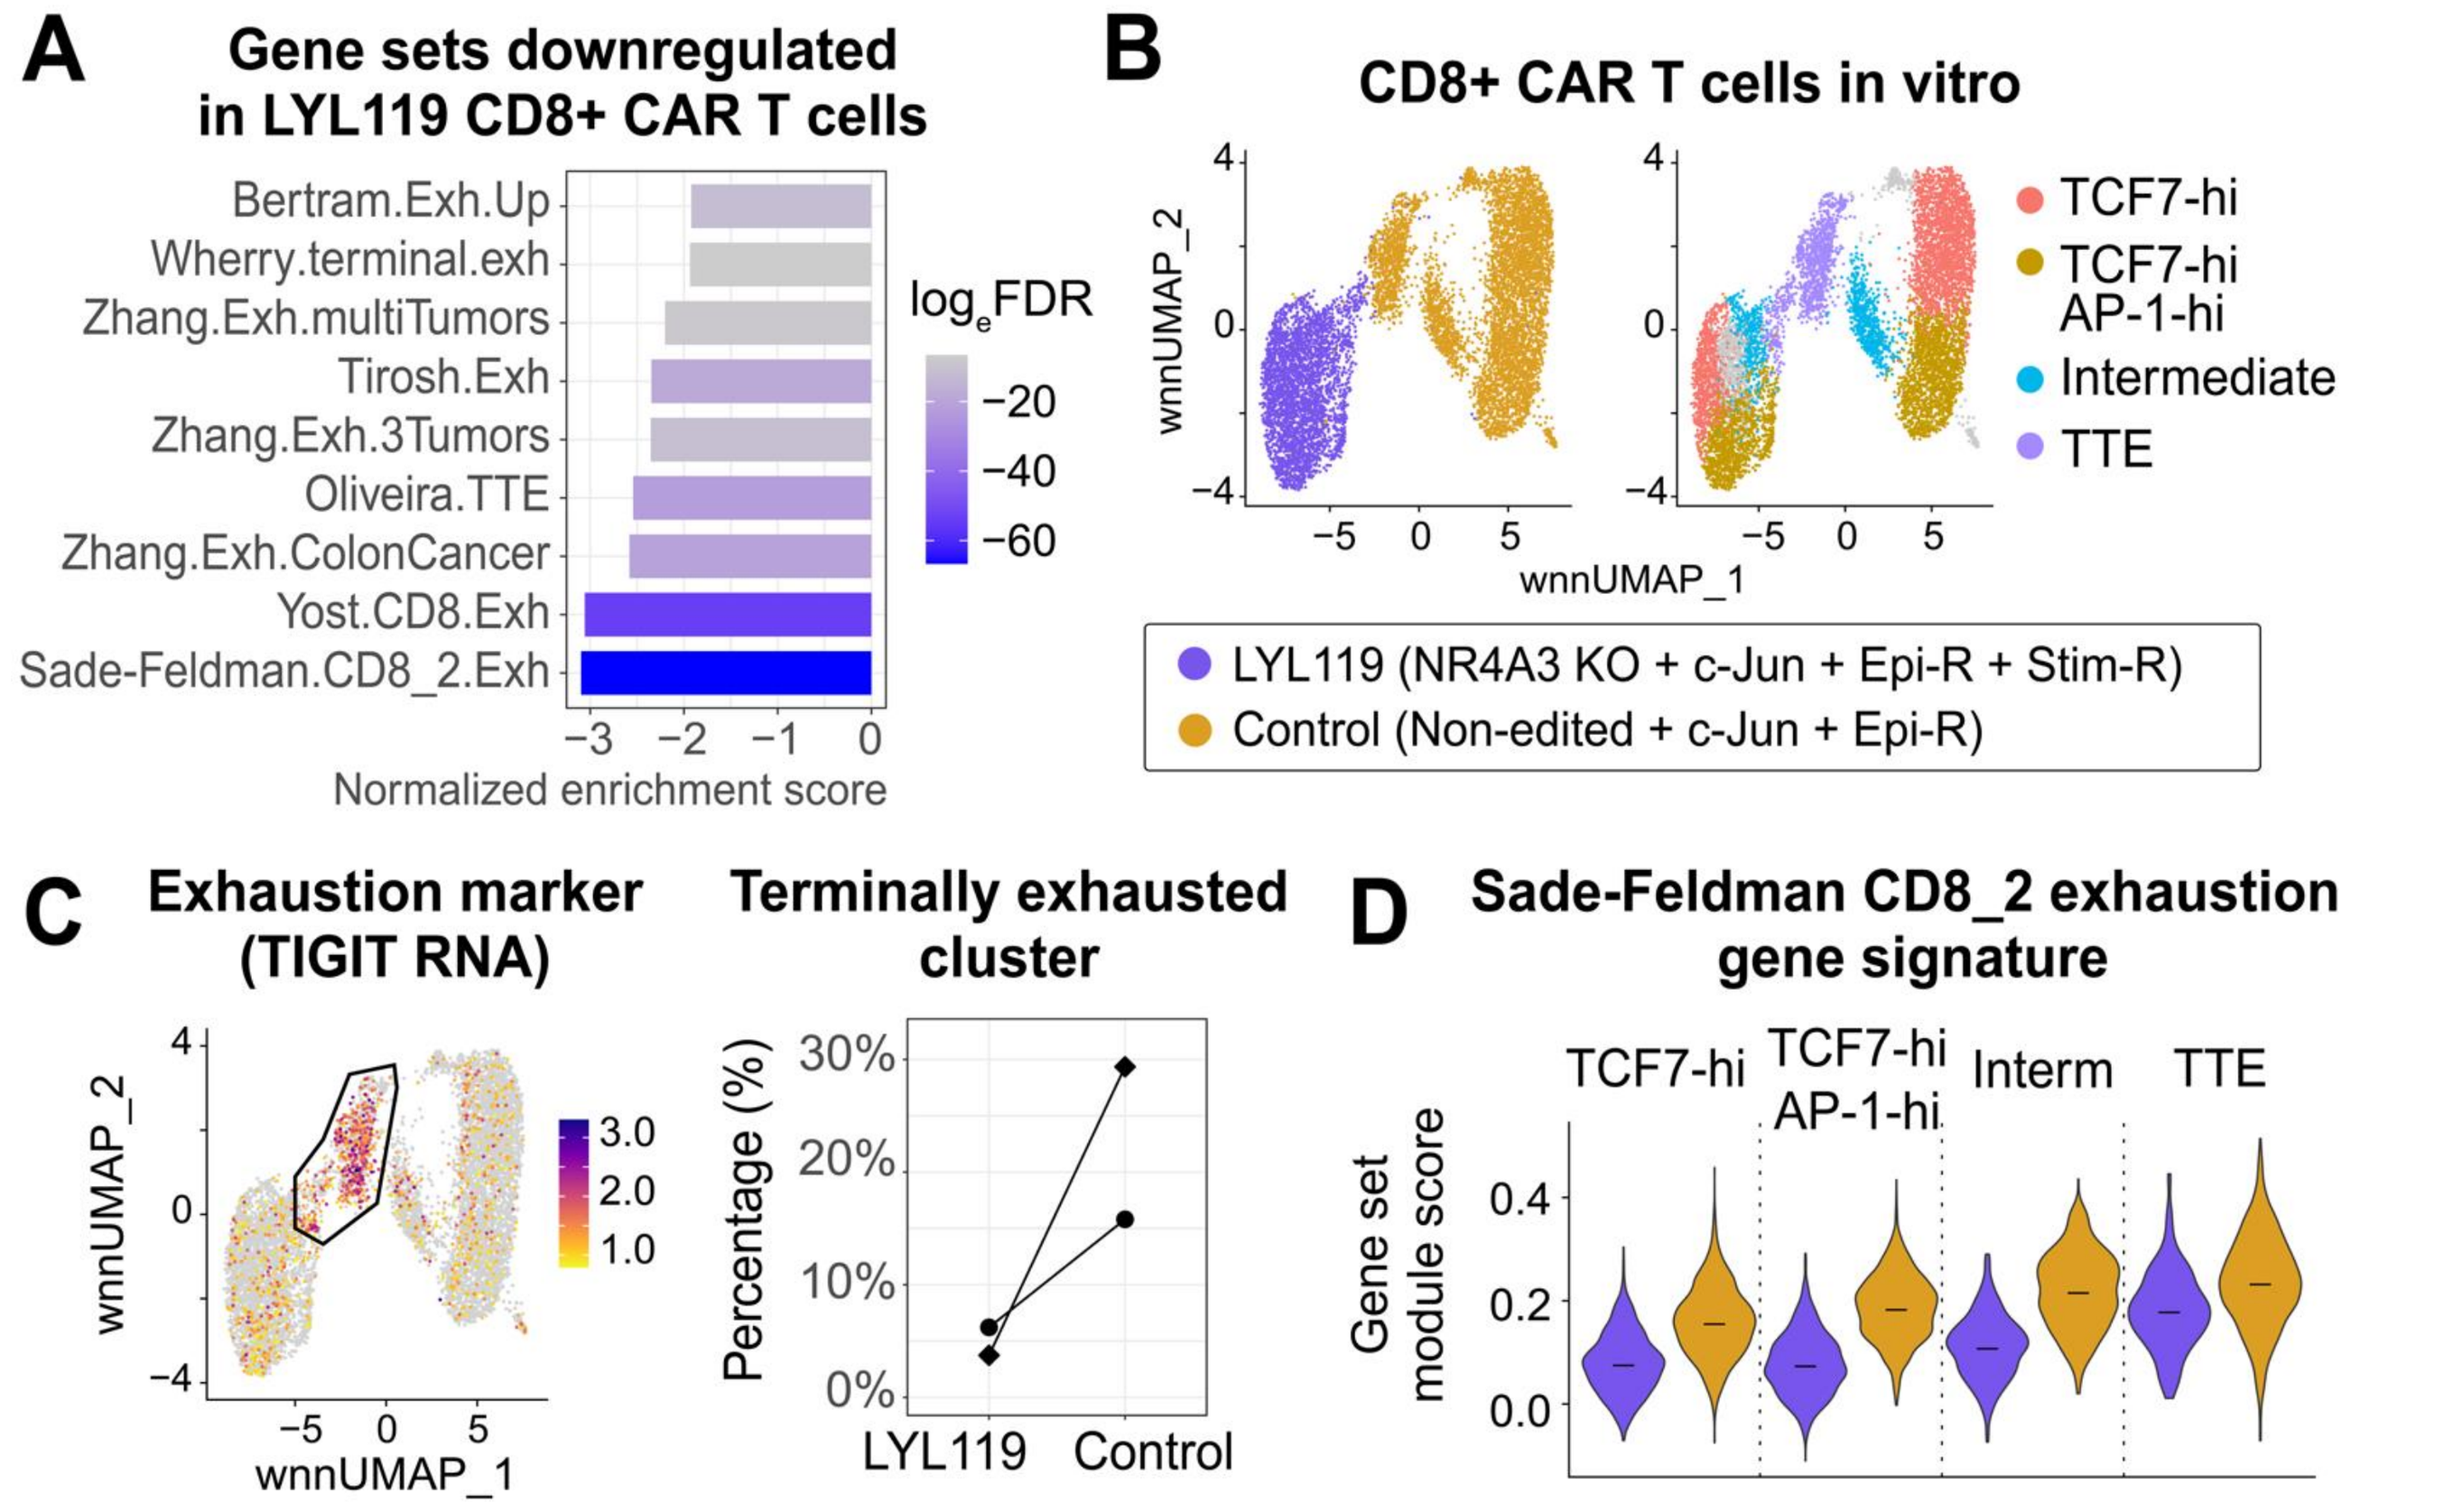

A
Gene sets downregulated in LYL119 CD8+ CAR T cells
Bertram.Exh.Up
Wherry.terminal.exh
Zhang.Exh.multiTumors
Tirosh.Exh
Zhang.Exh.3Tumors
Oliveira.TTE
Zhang.Exh.ColonCancer
Yost.CD8.Exh
Sade-Feldman.CD8_2.Exh
Normalized enrichment score
log_e FDR
B
CD8+ CAR T cells in vitro
wnnUMAP_1
wnnUMAP_2
TCF7-hi
TCF7-hi AP-1-hi
Intermediate
TTE
LYL119 (NR4A3 KO + c-Jun + Epi-R + Stim-R)
Control (Non-edited + c-Jun + Epi-R)
C
Exhaustion marker (TIGIT RNA)
Terminally exhausted cluster
Percentage (%)
LYL119
Control
D
Sade-Feldman CD8_2 exhaustion gene signature
Gene set module score
TCF7-hi
TCF7-hi AP-1-hi
Interm
TTE

(A) Exhaustion-related gene sets are downregulated in LYL119 compared to control CAR T cells in pseudobulk gene set enrichment analysis based on data from two representative products after serial restimulation. (B) UMAP plot showing phenotypic annotation of cell subsets found by unsupervised clustering. (C) The terminally exhausted cluster enriched for TIGIT RNA expression is circled and is present at a lower proportion in LYL119. Symbols represent independent products. (D) The CD8_2 exhaustion gene set[4] is downregulated in LYL119 across phenotypic cell subsets. Data from one representative product after serial restimulation is shown in (B), left panel of (C), and (D).

Figure 5: LYL119 exhibits decreased NR4A motif accessibility and reduced chromatin accessibility near exhaustion-related genes

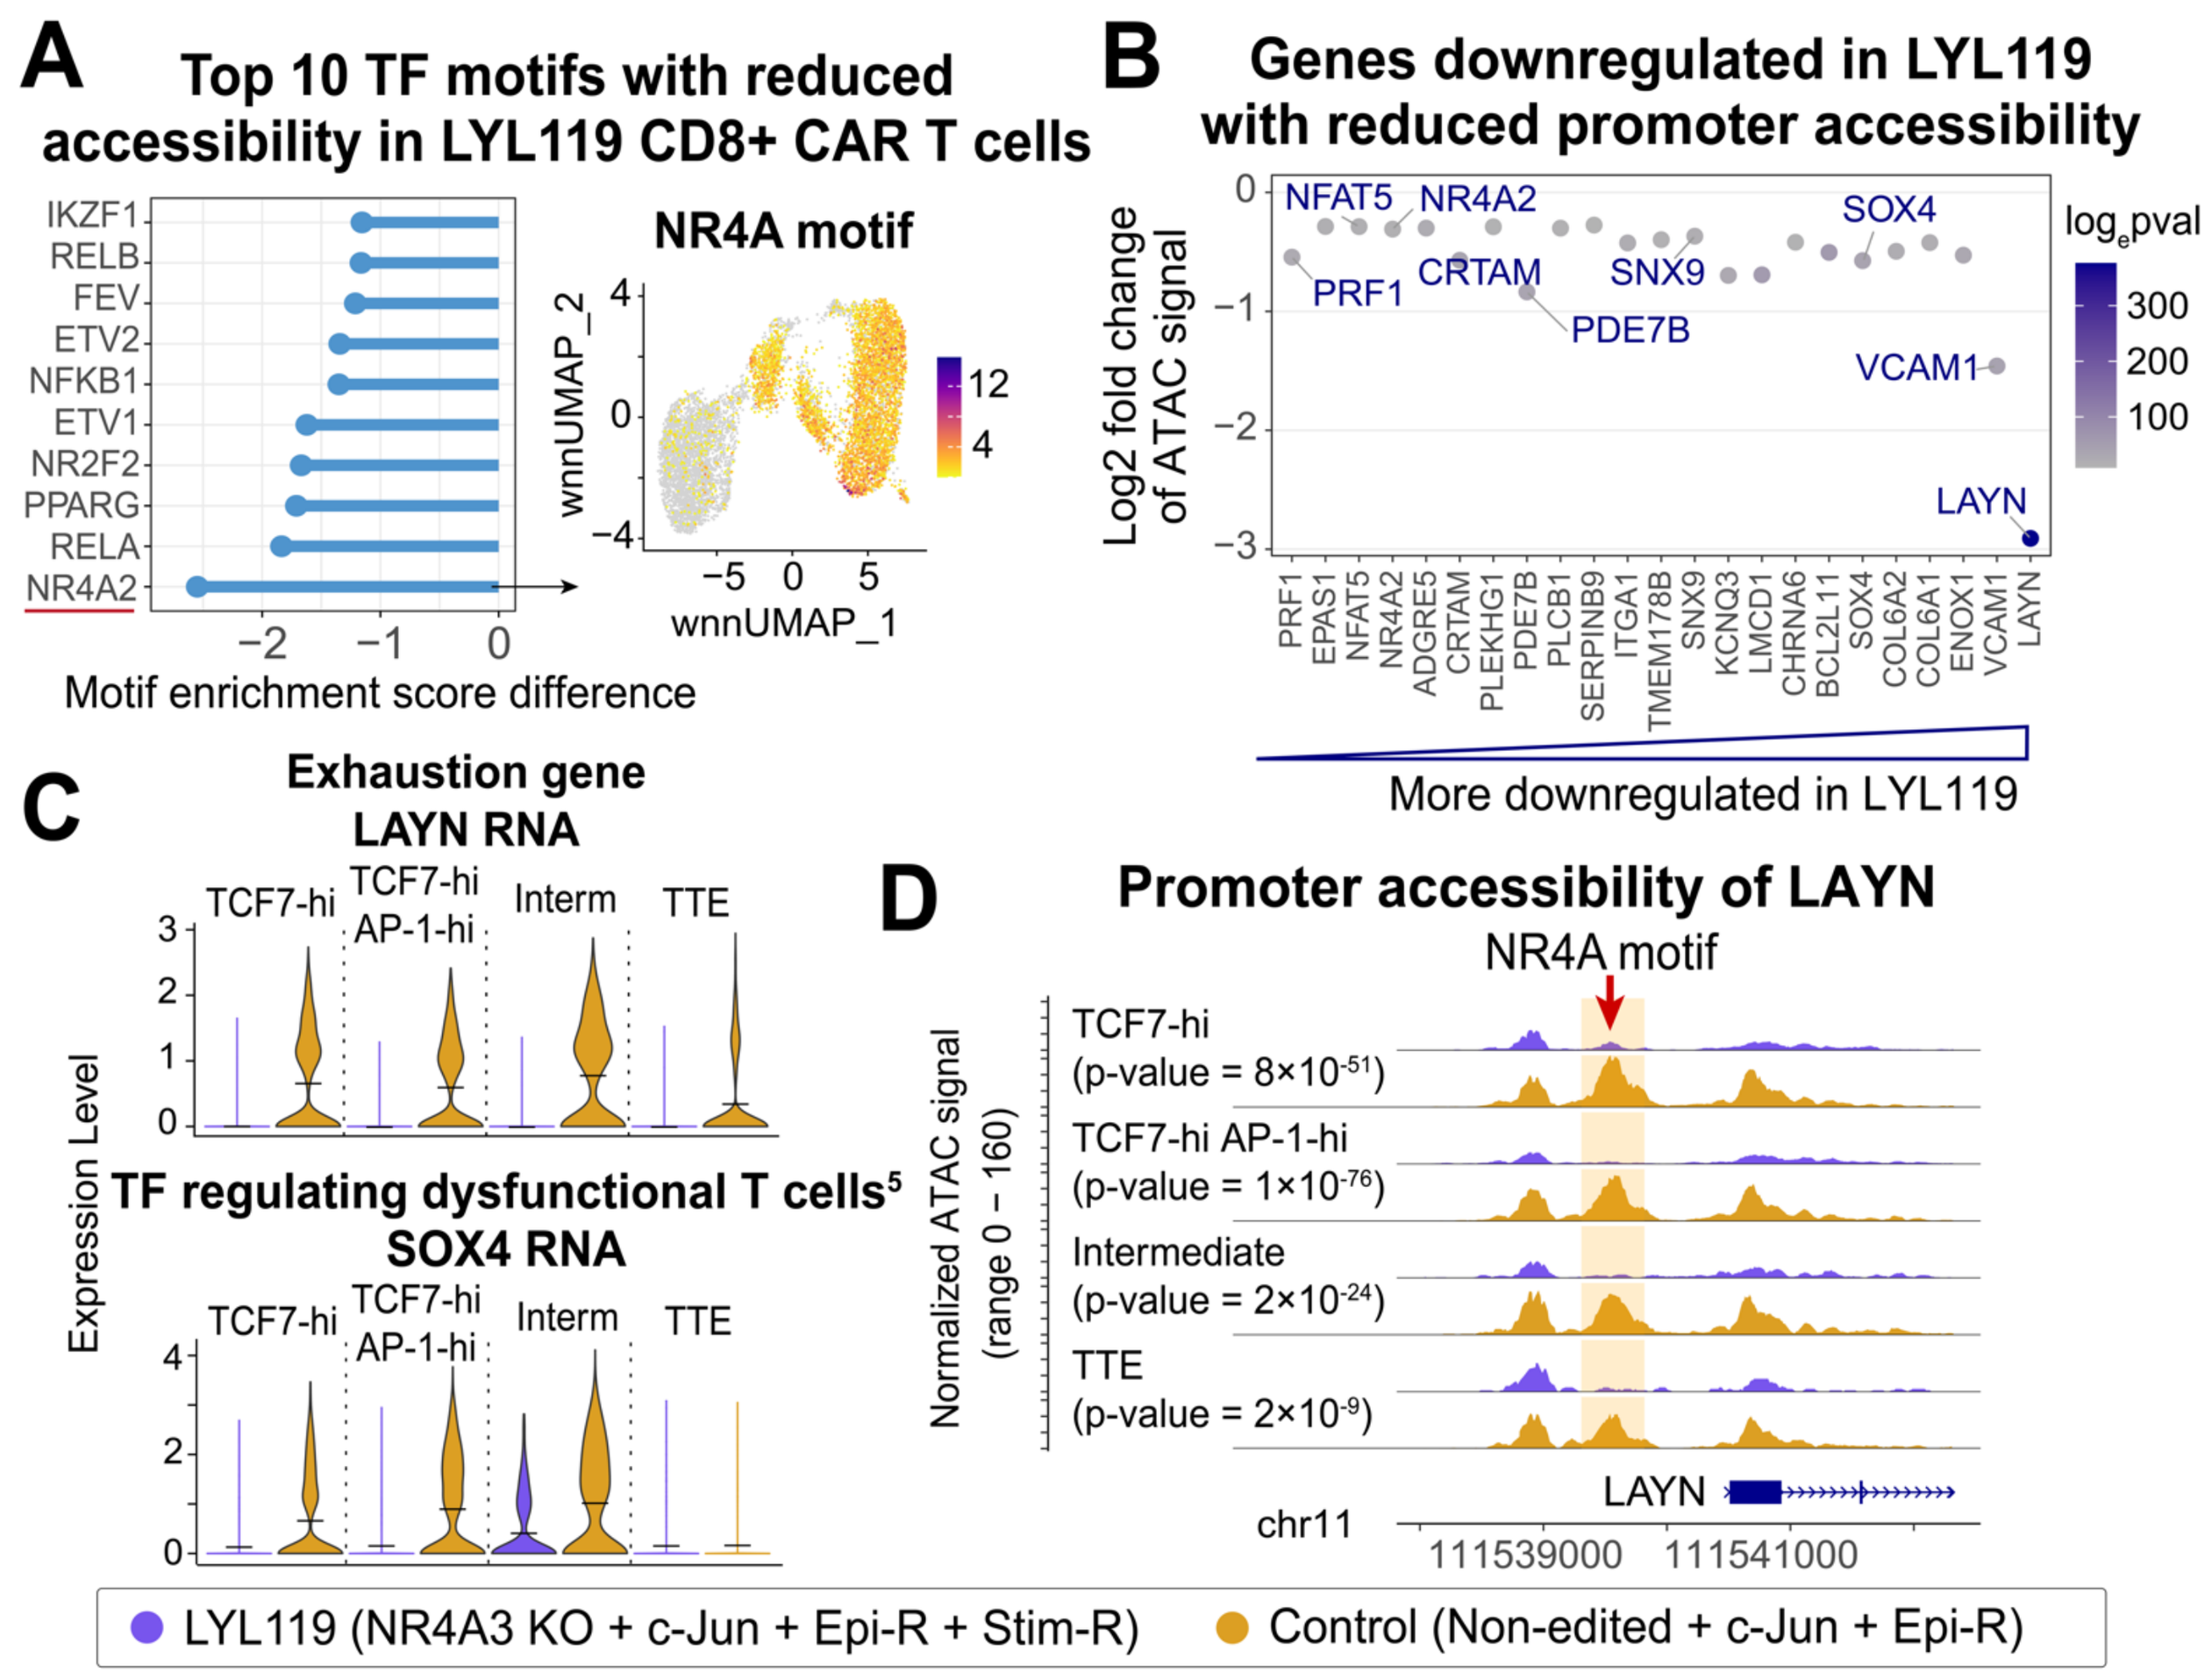

A
Top 10 TF motifs with reduced accessibility in LYL119 CD8+ CAR T cells
IKZF1
RELB
FEV
ETV2
NFKB1
ETV1
NR2F2
PPARG
RELA
NR4A2
Motif enrichment score difference
NR4A motif
wnnUMAP_1
wnnUMAP_2
B
Genes downregulated in LYL119 with reduced promoter accessibility
Log2 fold change of ATAC signal
NFAT5
NR4A2
PRF1
CRTAM
SNX9
PDE7B
SOX4
VCAM1
LAYN
log_e pval
More downregulated in LYL119
C
Exhaustion gene LAYN RNA
TF regulating dysfunctional T cells⁵ SOX4 RNA
Expression Level
TCF7-hi
TCF7-hi AP-1-hi
Interm
TTE
D
Promoter accessibility of LAYN
NR4A motif
Normalized ATAC signal (range 0 – 160)
TCF7-hi (p-value = 8×10⁻⁵¹)
TCF7-hi AP-1-hi (p-value = 1×10⁻⁷⁶)
Intermediate (p-value = 2×10⁻²⁴)
TTE (p-value = 2×10⁻⁹)
chr11
111539000
111541000
LYL119 (NR4A3 KO + c-Jun + Epi-R + Stim-R)
Control (Non-edited + c-Jun + Epi-R)

(A) Top 10 transcription factor (TF) motifs with reduced enrichment score in LYL119 compared to control CAR T cells. The NR4A motif (named by NR4A2) is highlighted. (B) Genes significantly downregulated in LYL119 with reduced promoter accessibility. Exhaustion-related genes from gene sets shown in Figure 4A are highlighted. (C) LAYN and SOX4 RNA expression. (D) NR4A motif is present in LAYN's promoter peak (highlighted) with significantly reduced accessibility in LYL119 across phenotypic cell subsets. Data from one representative product after serial restimulation is shown in (A), (C), and (D).

Figure 6: LYL119 exhibits enhanced memory-related gene expression associated with increased AP-1 motif accessibility and contains a TCF7-hi subset exhibiting effector-related characteristics

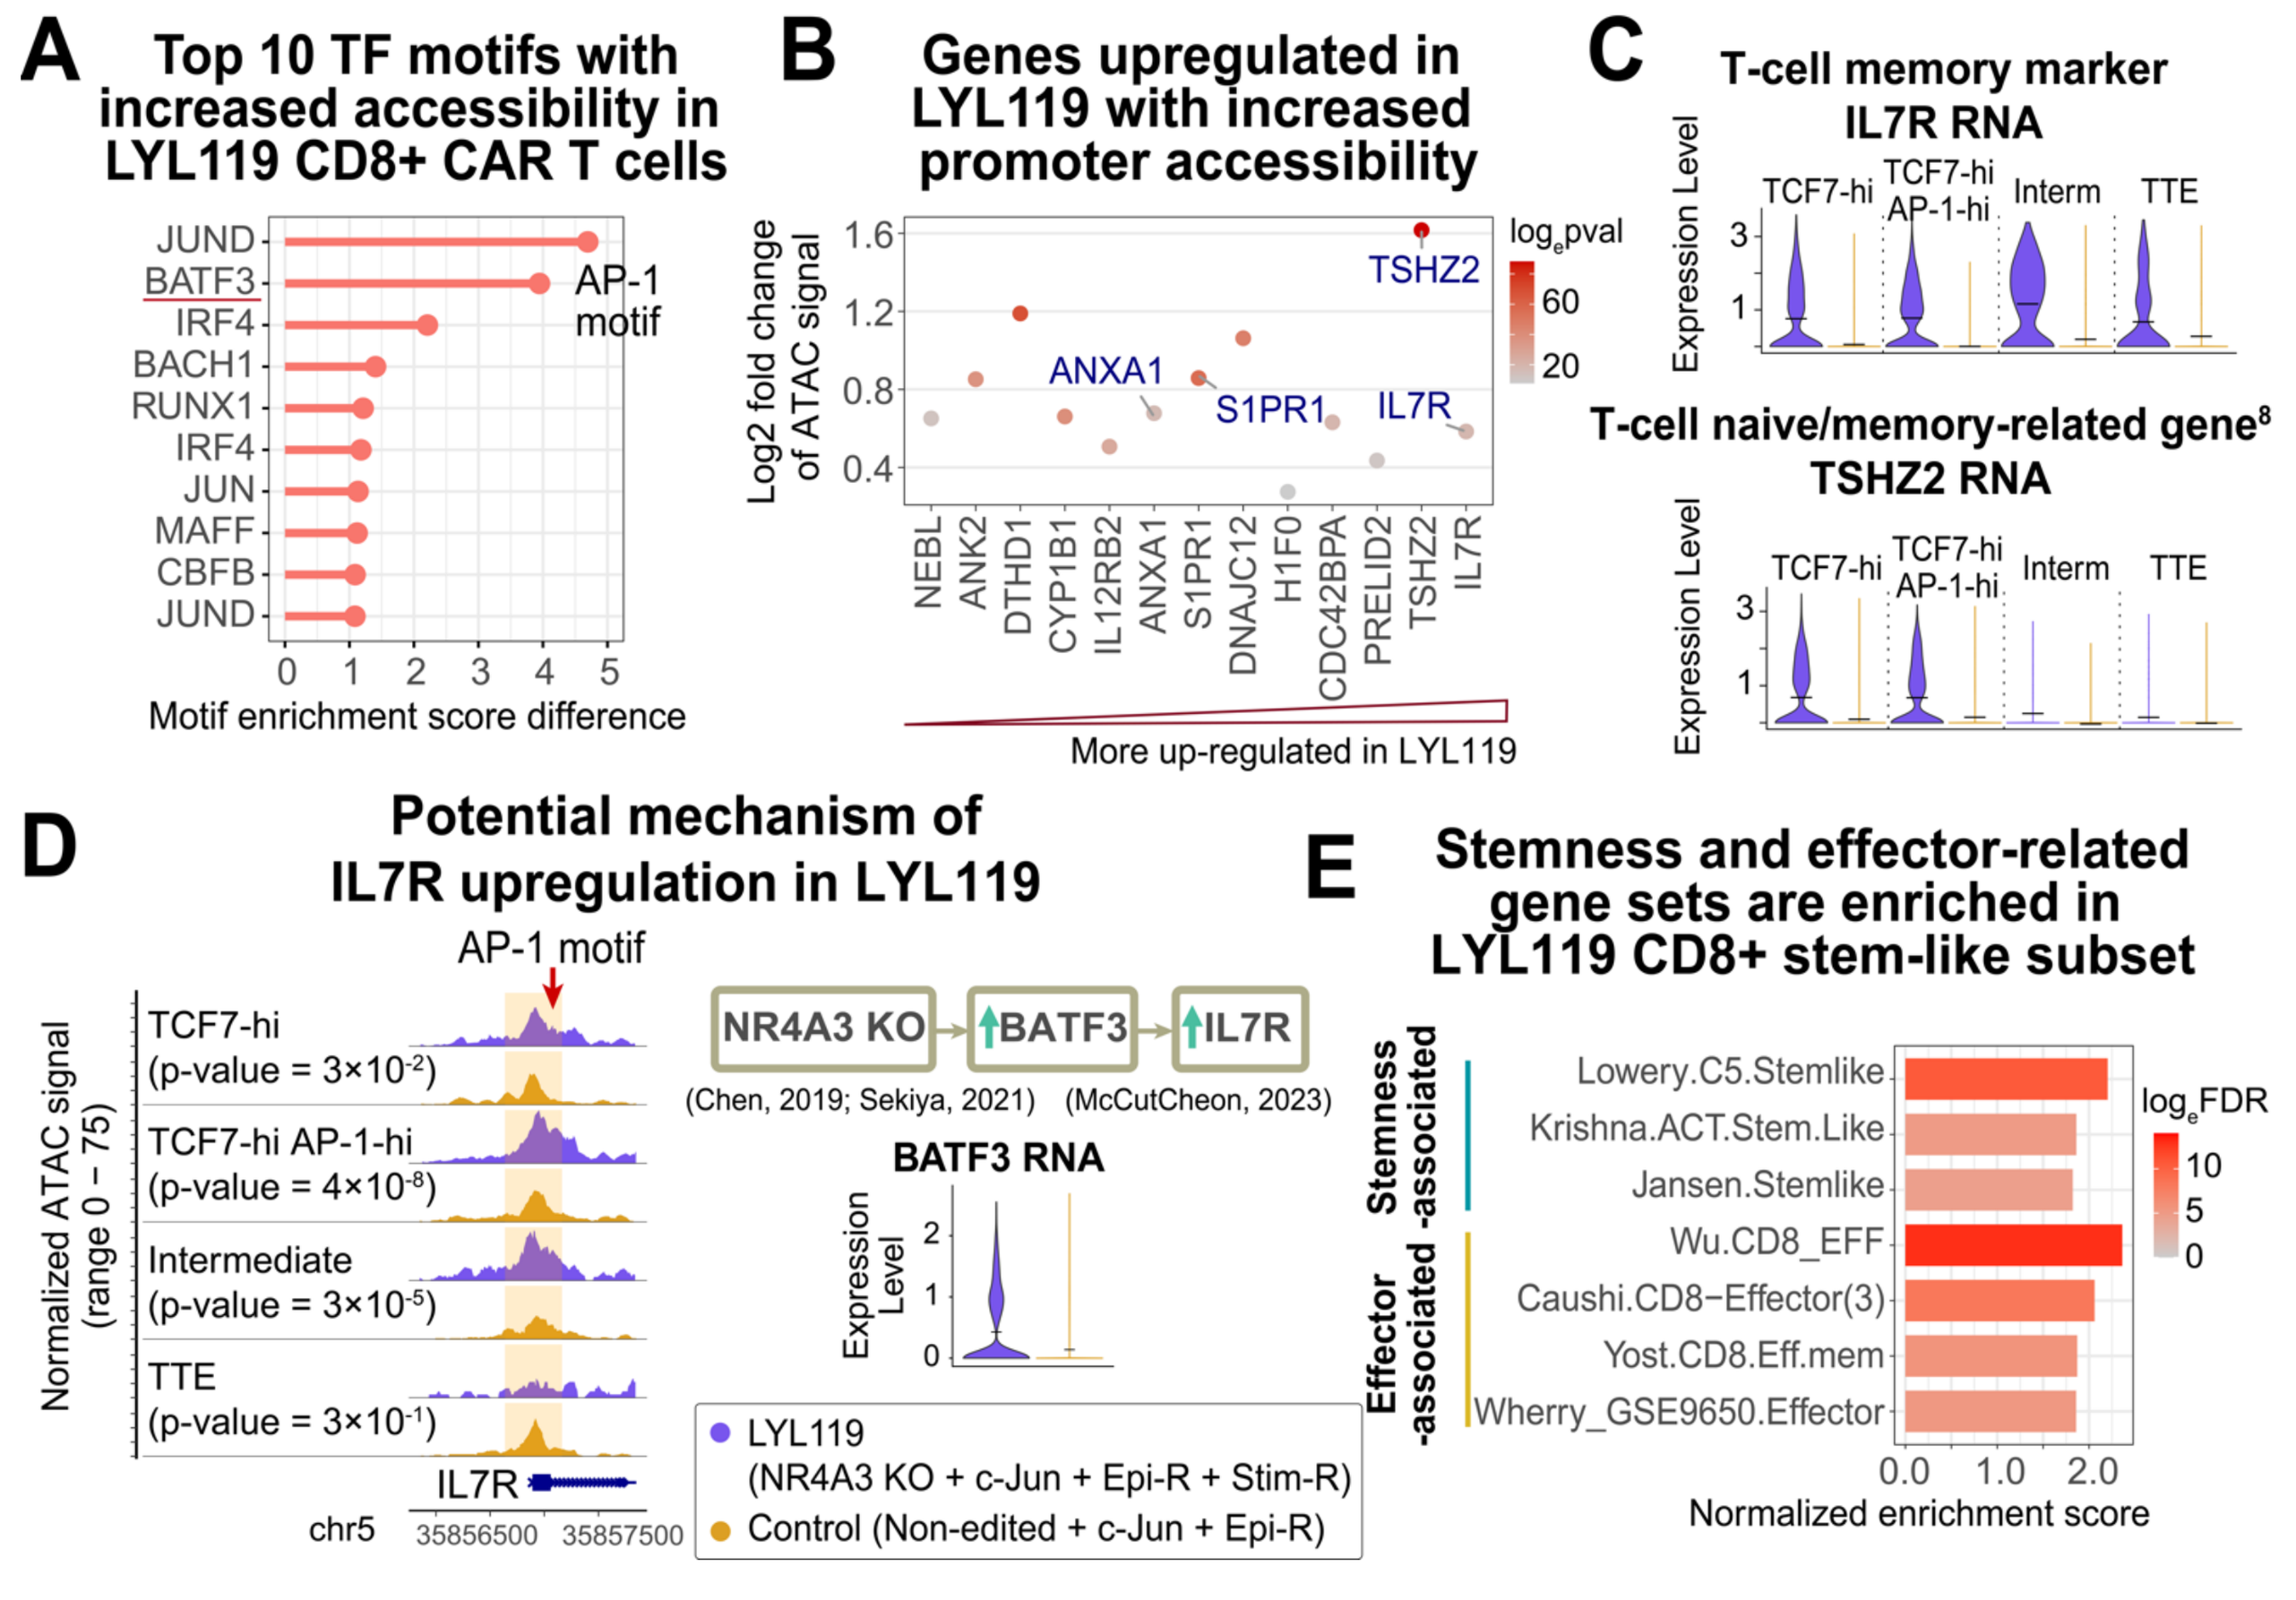

A
Top 10 TF motifs with increased accessibility in LYL119 CD8+ CAR T cells
JUND
BATF3
IRF4
BACH1
RUNX1
IRF4
JUN
MAFF
CBFB
JUND
AP-1 motif
Motif enrichment score difference
B
Genes upregulated in LYL119 with increased promoter accessibility
Log2 fold change of ATAC signal
TSHZ2
ANXA1
S1PR1
IL7R
log_e pval
More up-regulated in LYL119
C
T-cell memory marker IL7R RNA
T-cell naive/memory-related gene⁸ TSHZ2 RNA
Expression Level
TCF7-hi
TCF7-hi AP-1-hi
Interm
TTE
D
Potential mechanism of IL7R upregulation in LYL119
AP-1 motif
Normalized ATAC signal (range 0 – 75)
TCF7-hi (p-value = 3×10⁻²)
TCF7-hi AP-1-hi (p-value = 4×10⁻⁸)
Intermediate (p-value = 3×10⁻⁵)
TTE (p-value = 3×10⁻¹)
chr5
IL7R
NR4A3 KO → ↑BATF3 → ↑IL7R
(Chen, 2019; Sekiya, 2021) (McCutCheon, 2023)
BATF3 RNA
LYL119 (NR4A3 KO + c-Jun + Epi-R + Stim-R)
Control (Non-edited + c-Jun + Epi-R)
E
Stemness and effector-related gene sets are enriched in LYL119 CD8+ stem-like subset
Stemness -associated
Effector -associated
Lowery.C5.Stemlike
Krishna.ACT.Stem.Like
Jansen.Stemlike
Wu.CD8_EFF
Caushi.CD8−Effector(3)
Yost.CD8.Eff.mem
Wherry_GSE9650.Effector
log_e FDR
Normalized enrichment score

(A) Top 10 TF motifs with increased enrichment score in LYL119 compared to control CAR T cells. The AP-1 motif (named by BATF3) is highlighted. (B) Genes significantly upregulated in LYL119 with increased promoter accessibility. Stemness-related genes[6-7] are highlighted. (C) IL7R and TSHZ2 RNA expression. (D) AP-1 motif is present in the promoter peak of IL7R (highlighted) with increased accessibility in LYL119. One potential regulatory mechanism is shown. (E) The TCF7-hi stem-like subset in LYL119 shows elevated stemness-related and effector-related gene signatures compared to its counterpart in control. Data from one representative product after serial restimulation is shown in (A), (C), and (D).

LYL119 exhibits an improved phenotype in an in vivo xenograft tumor model compared to control (Figure 7)

On day 14 after T-cell injection (1 x 10^6 CAR T-cell dose), tumor-infiltrating ROR1 CAR+ T cells from LYL119 exhibits:

- Downregulation of exhaustion-related gene signatures and lower proportion of terminally exhausted T cells
- Upregulation of memory/stemness-related gene signatures
- Lower proportion of a potentially suppressive FOXP3-hi T-cell cluster

Figure 7: LYL119 exhibits reduced exhaustion and enhanced T-cell memory-related gene signatures in vivo

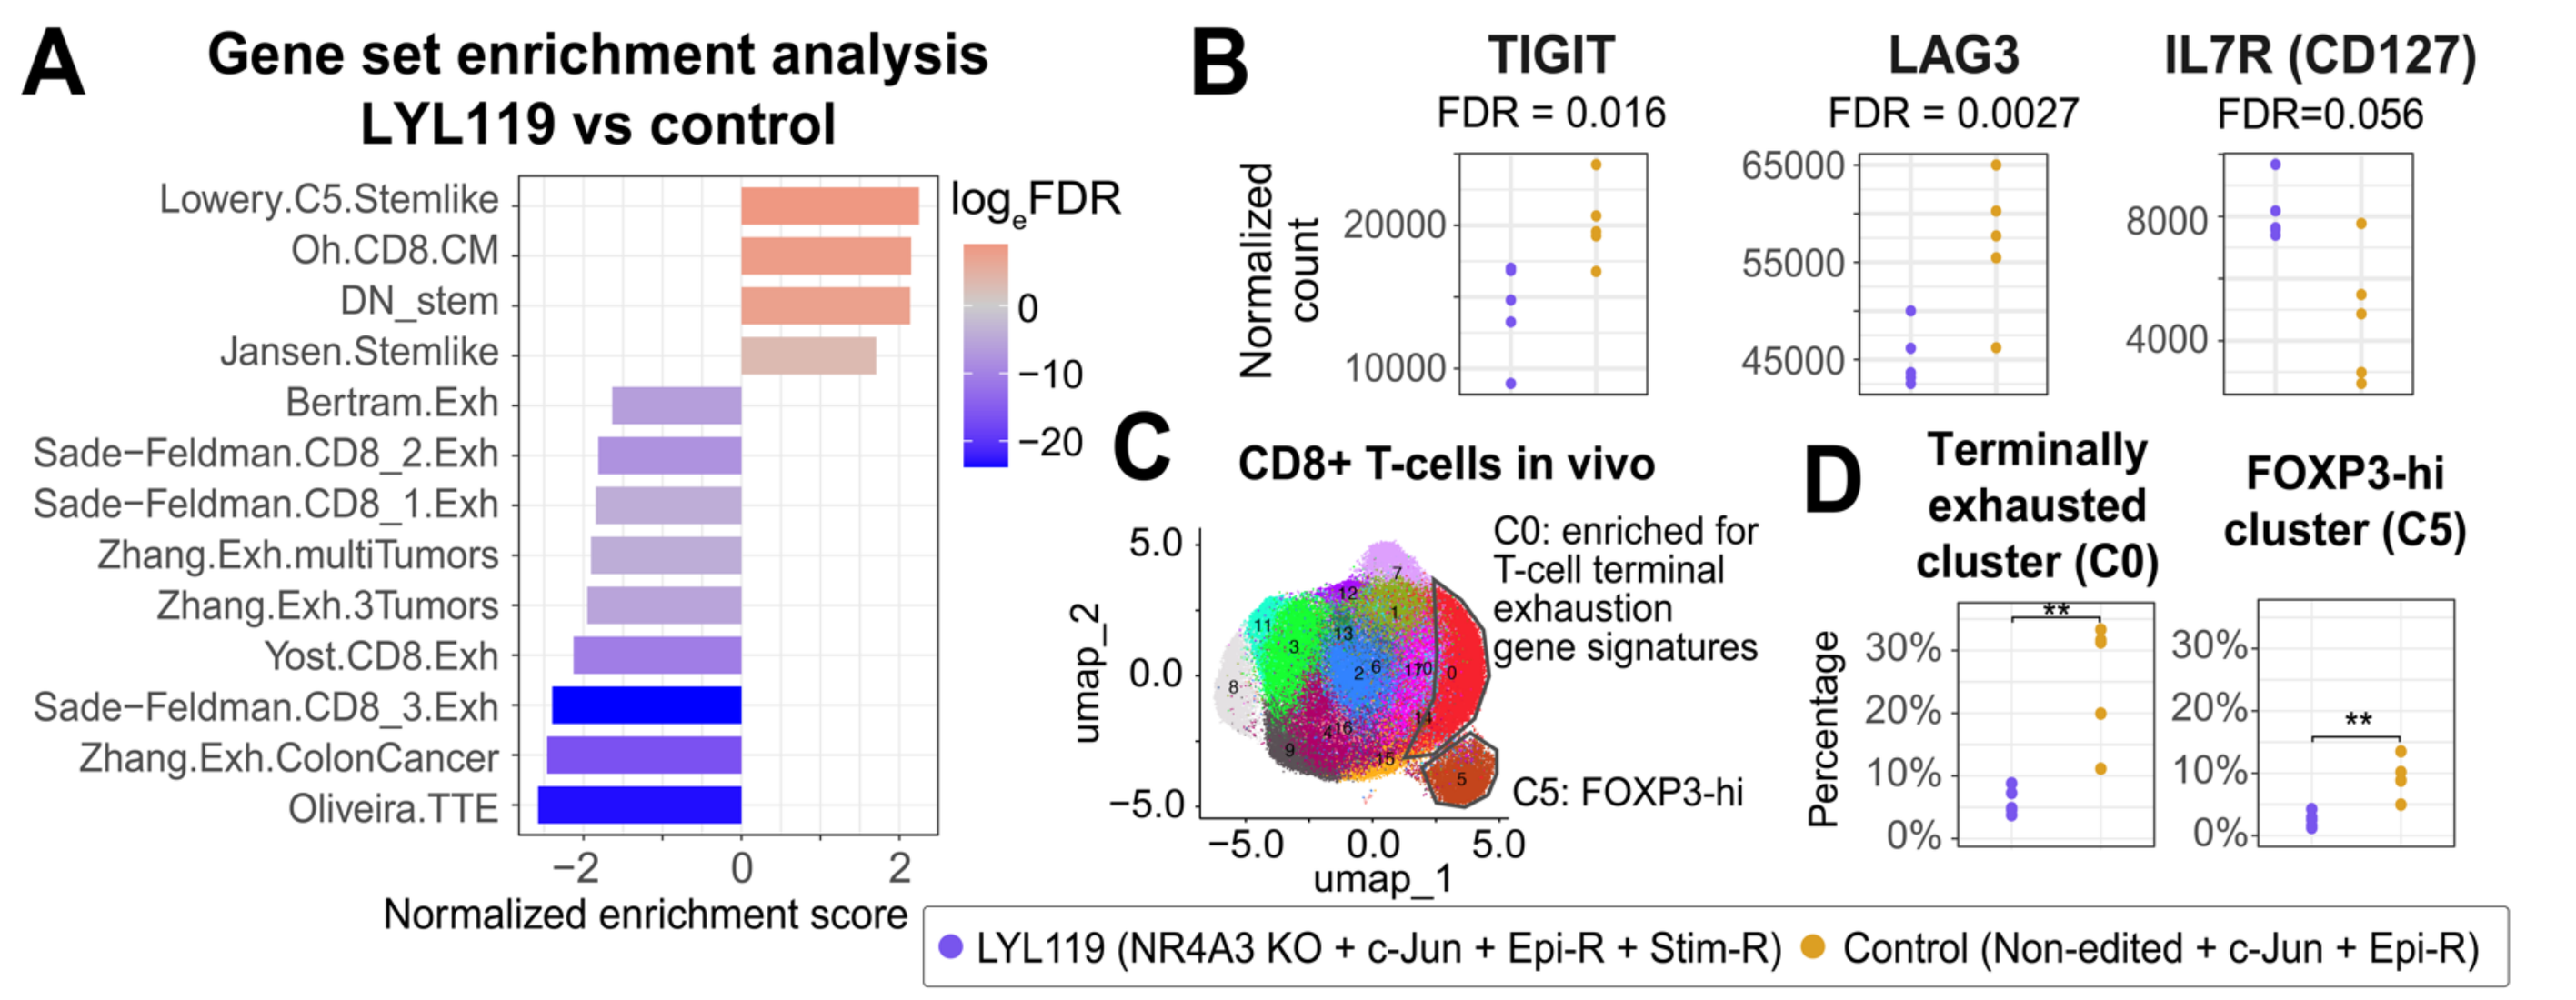

A
Gene set enrichment analysis LYL119 vs control
Lowery.C5.Stemlike
Oh.CD8.CM
DN_stem
Jansen.Stemlike
Bertram.Exh
Sade−Feldman.CD8_2.Exh
Sade−Feldman.CD8_1.Exh
Zhang.Exh.multiTumors
Zhang.Exh.3Tumors
Yost.CD8.Exh
Sade−Feldman.CD8_3.Exh
Zhang.Exh.ColonCancer
Oliveira.TTE
Normalized enrichment score
log_e FDR
B
TIGIT FDR = 0.016
LAG3 FDR = 0.0027
IL7R (CD127) FDR=0.056
Normalized count
C
CD8+ T-cells in vivo
C0: enriched for T-cell terminal exhaustion gene signatures
C5: FOXP3-hi
umap_1
umap_2
D
Terminally exhausted cluster (C0)
FOXP3-hi cluster (C5)
Percentage
LYL119 (NR4A3 KO + c-Jun + Epi-R + Stim-R)
Control (Non-edited + c-Jun + Epi-R)

(A) Pseudobulk gene set enrichment analysis results of exhaustion-related gene sets and T-cell stemness/memory-related gene sets based on data from 10 mice. (B) RNA expression of TIGIT, LAG3 and IL7R. (C) UMAP plot highlighting the cluster most enriched for T-cell terminal exhaustion gene signatures (C0) and a FOXP3-hi cluster (C5). (D) Proportion of C0 and C5. Symbols represent individual mice in each treatment group. Asterisks indicate significant differences.

Conclusions

LYL119, a ROR1-targeted CAR T-cell product enhanced with c-Jun overexpression, NR4A3 KO, Epi-R manufacturing protocol, and Stim-R technology exhibits:

- Superior in vitro activity compared to control CAR T cells and antitumor activity in vivo even at the low dose of 0.1 x 10^6 cells.
- Reduced T-cell exhaustion and enhanced memory-related characteristics after antigen encounter in vitro and in vivo.
- Reduced accessibility of the NR4A binding motif and increased accessibility of the AP-1 binding motif in vitro.

These data suggest that enhanced LYL119 antitumor functions may be attributed to reduction of T-cell exhaustion and maintenance of T-cell memory characteristics. The improved phenotype potentially results from epigenetic changes favoring AP-1 family transcription factor binding while reducing NR4A binding. LYL119 is entering Phase I clinical development in patients with ROR1+ solid tumors and will initially enroll patients with platinum-resistant ovarian cancer or relapsed/refractory endometrial cancer.

Abbreviations:
AUC, area under the curve; AP-1, activator protein 1; BATF3: basic leucine zipper transcriptional factor ATF-like 3; CAR, chimeric antigen receptor; CD, cluster of differentiation; dKO, double knockout; E:T, effector-to-target; FOXP3, forkhead box protein P3; IFN-γ, interferon gamma; IL-2, interleukin 2; IL7R, interleukin 7 receptor; Interm, Intermediate; KO, knockout; LAG3, lymphocyte activation gene 3; LAYN, layilin; NR4A3, nuclear receptor subfamily 4 group A member 3; MHC, major histocompatibility complex; NSCLC, non-small cell lung cancer; NSG, NOD scid gamma; ROR1, receptor tyrosine kinase-like orphan receptor 1; SD, standard deviation; SEM, standard error of the mean; SOX4, SRY-box transcription factor 4; TCF7, transcription factor 7; TF, transcription factor; TIGIT, T cell immunoreceptor with Ig and ITIM domains. TSHZ2, teashirt zinc finger homeobox 2.

* $p < 0.05$; ** $p < 0.01$; *** $p < 0.001$; **** $p < 0.0001$.

References
1. Albelda SM. Nat Rev Clin Oncol. 2024;21(1):47-66
2. Krishna S, et al. *Science*. 2020;370:1328–1334
3. Lam V, et al. SITC 2023 Poster. Abstract 278
4. Sade-Feldman M, et al. Cell. 2018;175(4):998-1013.e20
5. Good CR, et al. Cell. 2021;184(25):6081-6100.e26
6. Caushi JX, et al. Nature. 2021;596(7870):126-132
7. Jansen CS, et al. Nature. 2019;576(7787):465-470
8. Zhao, J, et al. Cell discovery. 2020; 6(1): 22

Acknowledgement
We thank members of Lyell Immunopharma's In Vivo Pharmacology team (Ally Torres Diaz, Eskedar Nigatu, and Nik Vad), NGS/Flow team (Emily Fu-Sum, Smita Ghanekar, Sheila Lou, and Taran Ramadoss), Research team (Omar Ali, Jessica Briones, Alexander S. Cheung, and Aileen Li), Process Sciences team (Meritxell Galindo Casas, Esha Afreen, Grant Vavra) and Vector Sciences team (David Anderson and Martin Wohlfahrt) for their experimental contributions.

©2024 Lyell Immunopharma, Inc.