NR4A3 gene editing and c-Jun overexpression synergize to limit exhaustion and enhance functional activity of ROR1 CAR T cells *in vitro* and *in vivo*

Viola C. Lam, Jessica Barragan, Christina Cheung, Jia Lu, David Chian, Rowena Martinez, Candace Sims, Purnima Sundar, Hajime Hiraragi, Shobha Potluri, Rachel C. Lynn

Lyell Immunopharma, 201 Haskins Way, South San Francisco, CA 94080.

Abstract No. 243

Background

T-cell exhaustion limits the efficacy and durability of CAR T-cell therapies in solid tumors

- CAR T-cell therapy can have substantial clinical efficacy in patients with hematologic malignancies, but responses have thus far been limited in solid tumors[1]
- Solid tumors pose many barriers to successful T-cell therapy, including the development of cell-intrinsic dysfunction or exhaustion[2]
- Next-generation strategies to improve T-cell functional activity, persistence, and durability are needed for effective cellular immunotherapy against solid tumors

Genetic reprogramming to overexpress c-Jun can delay T-cell exhaustion

- Dysregulation of activator protein 1 (AP-1) family genes has been implicated in T-cell exhaustion[3]
- Overexpression of the AP-1 family transcription factor c-Jun reduces CAR T-cell exhaustion, thereby improving functional activity in multiple preclinical models[3]
- Application of this strategy through Lyell's genetic reprogramming technology to overexpress c-Jun prolongs effector function and delays exhaustion of ROR1 CAR T cells[4] (**Figure 1**)

Figure 1: Effects of c-Jun overexpression on T-cell activity

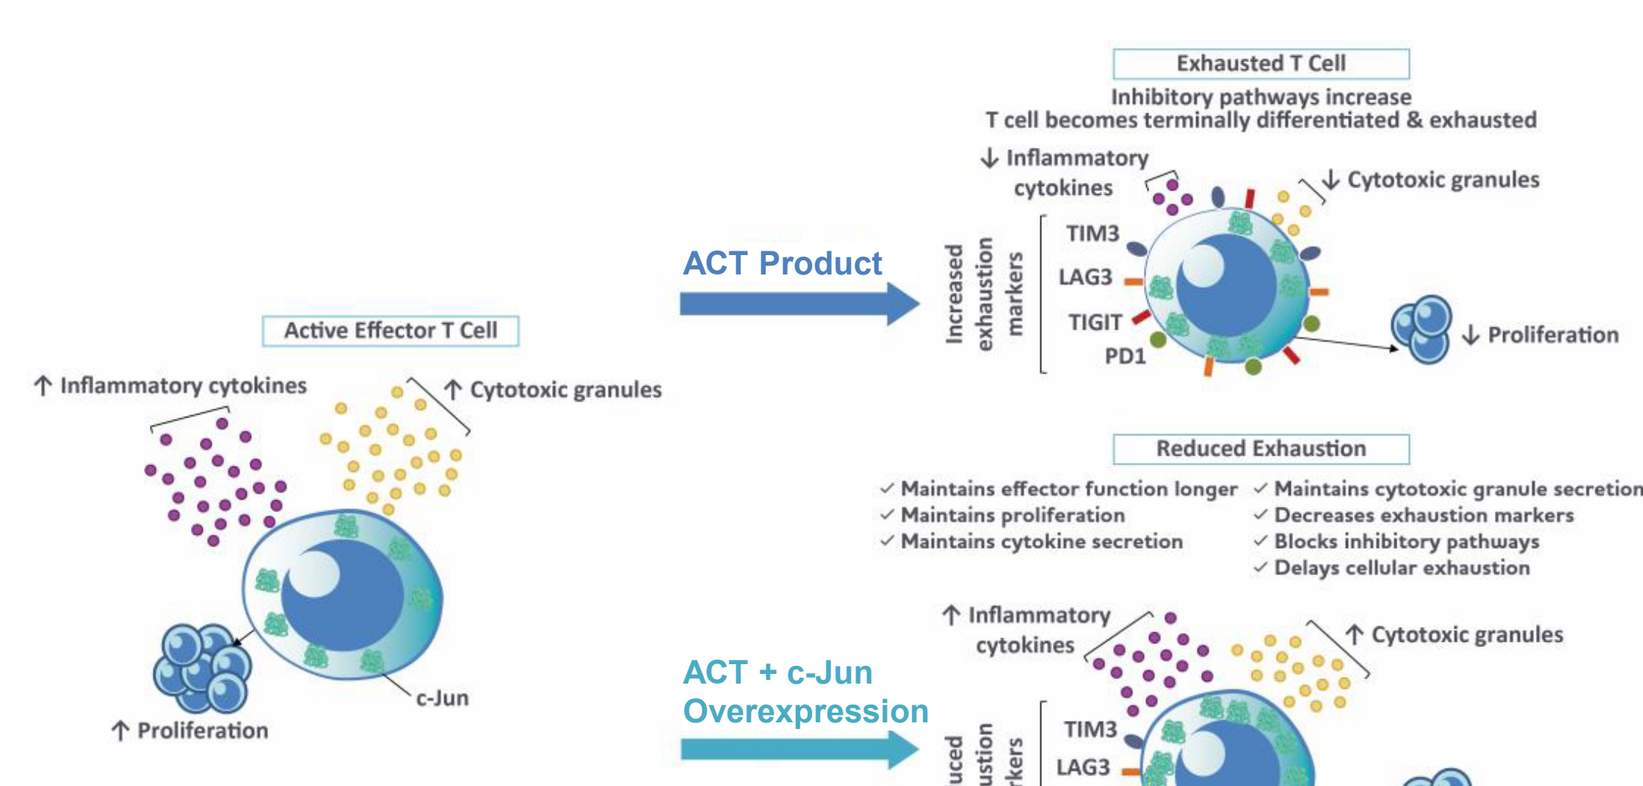
Active Effector T Cell
↑ Inflammatory cytokines
↑ Cytotoxic granules
↑ Proliferation
c-Jun
ACT Product
Exhausted T Cell
Inhibitory pathways increase
T cell becomes terminally differentiated & exhausted
↓ Inflammatory cytokines
↓ Cytotoxic granules
Increased exhaustion markers
TIM3
LAG3
TIGIT
PD1
↓ Proliferation
Reduced Exhaustion
✓ Maintains effector function longer
✓ Maintains proliferation
✓ Maintains cytokine secretion
✓ Maintains cytotoxic granule secretion
✓ Decreases exhaustion markers
✓ Blocks inhibitory pathways
✓ Delays cellular exhaustion
ACT + c-Jun Overexpression
↑ Inflammatory cytokines
↑ Cytotoxic granules
Reduced exhaustion markers
↑ Proliferation

NR4A family transcription factors regulate c-Jun activity in T cells

- NR4A family transcription factors may contribute to exhaustion and limit T-cell function by restraining activity of c-Jun
- The nuclear receptor subfamily 4A (NR4A) family of transcription factors (NR4A1, NR4A2, and NR4A3) are upregulated in exhausted CD8+ tumor-infiltrating lymphocytes (TIL)[5-7]
- Disruption of NR4A expression in adoptively transferred T cells can reduce dysfunction and enhance anti-tumor T-cell efficacy in murine tumor models[5,7]
- Reducing NR4A expression enhances sustained T-cell effector function associated with increased chromatin accessibility at AP-1 binding sites and increased expression of AP-1–regulated genes[5,7,8]

Hypothesis

We hypothesize that the combination of NR4A knockout (KO) and c-Jun overexpression may synergize to further limit exhaustion and enhance CAR T-cell function (Figure 2).

Figure 2: Effects of c-Jun and NR4A expression in T-cell activity

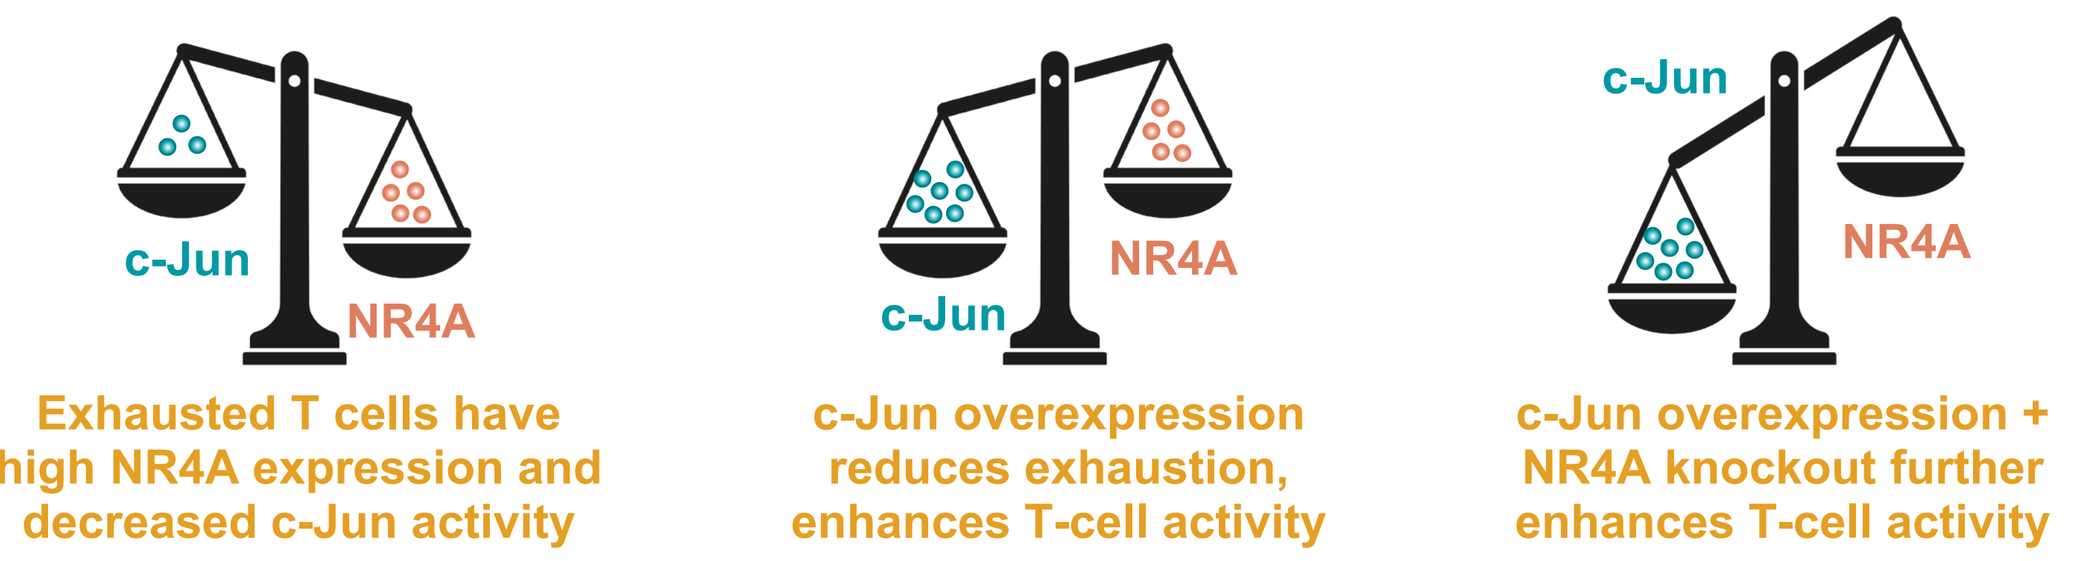
c-Jun
NR4A
Exhausted T cells have high NR4A expression and decreased c-Jun activity
c-Jun overexpression reduces exhaustion, enhances T-cell activity
c-Jun overexpression + NR4A knockout further enhances T-cell activity

Methods

- Healthy donor T cells were transduced with a ROR1 CAR lentiviral vector with or without c-Jun overexpression
- NR4A family genes (*NR4A1*, *NR4A2*, or *NR4A3*) were disrupted using CRISPR/Cas9 ribonucleoprotein (RNP) delivery via electroporation (EP)
- CAR T-cell cytotoxicity and cytokine production were evaluated *in vitro* after primary and repeated antigen-stimulation assays designed to promote exhaustion
- Cell phenotypes were assessed by flow cytometry and transcriptional profiling (bulk and single-cell RNA-Seq)
- CAR T cells were evaluated *in vivo* using a ROR1-expressing H1975 lung cancer xenograft model in mice

Results

NR4A3 KO results in superior CAR T-cell function compared to *NR4A1* or *NR4A2* KO

- To compare the relative effects of NR4A family transcription factor disruption on human CAR T-cell function, we designed CRISPR/Cas9 guide RNAs (gRNAs) targeting *NR4A1*, *NR4A2*, and *NR4A3*
- Activated T cells were transduced with a bi-cistronic human ROR1 CAR (ROR1 scFv-BBz—2A—EGFRt) lentiviral vector and electroporated 24 h later with Cas9/gRNA RNPs to produce NR4A KO or EP control ROR1 CAR T cells
- *NR4A3* KO ROR1 CAR T cells consistently demonstrated superior cytotoxic activity and prolonged cytokine production upon repeated antigen stimulation compared to *NR4A1* KO, *NR4A2* KO, and EP control ROR1 CAR T cells (**Figure 3**)

Figure 3: *NR4A3* KO enhances ROR1 CAR T-cell function *in vitro* and *in vivo*

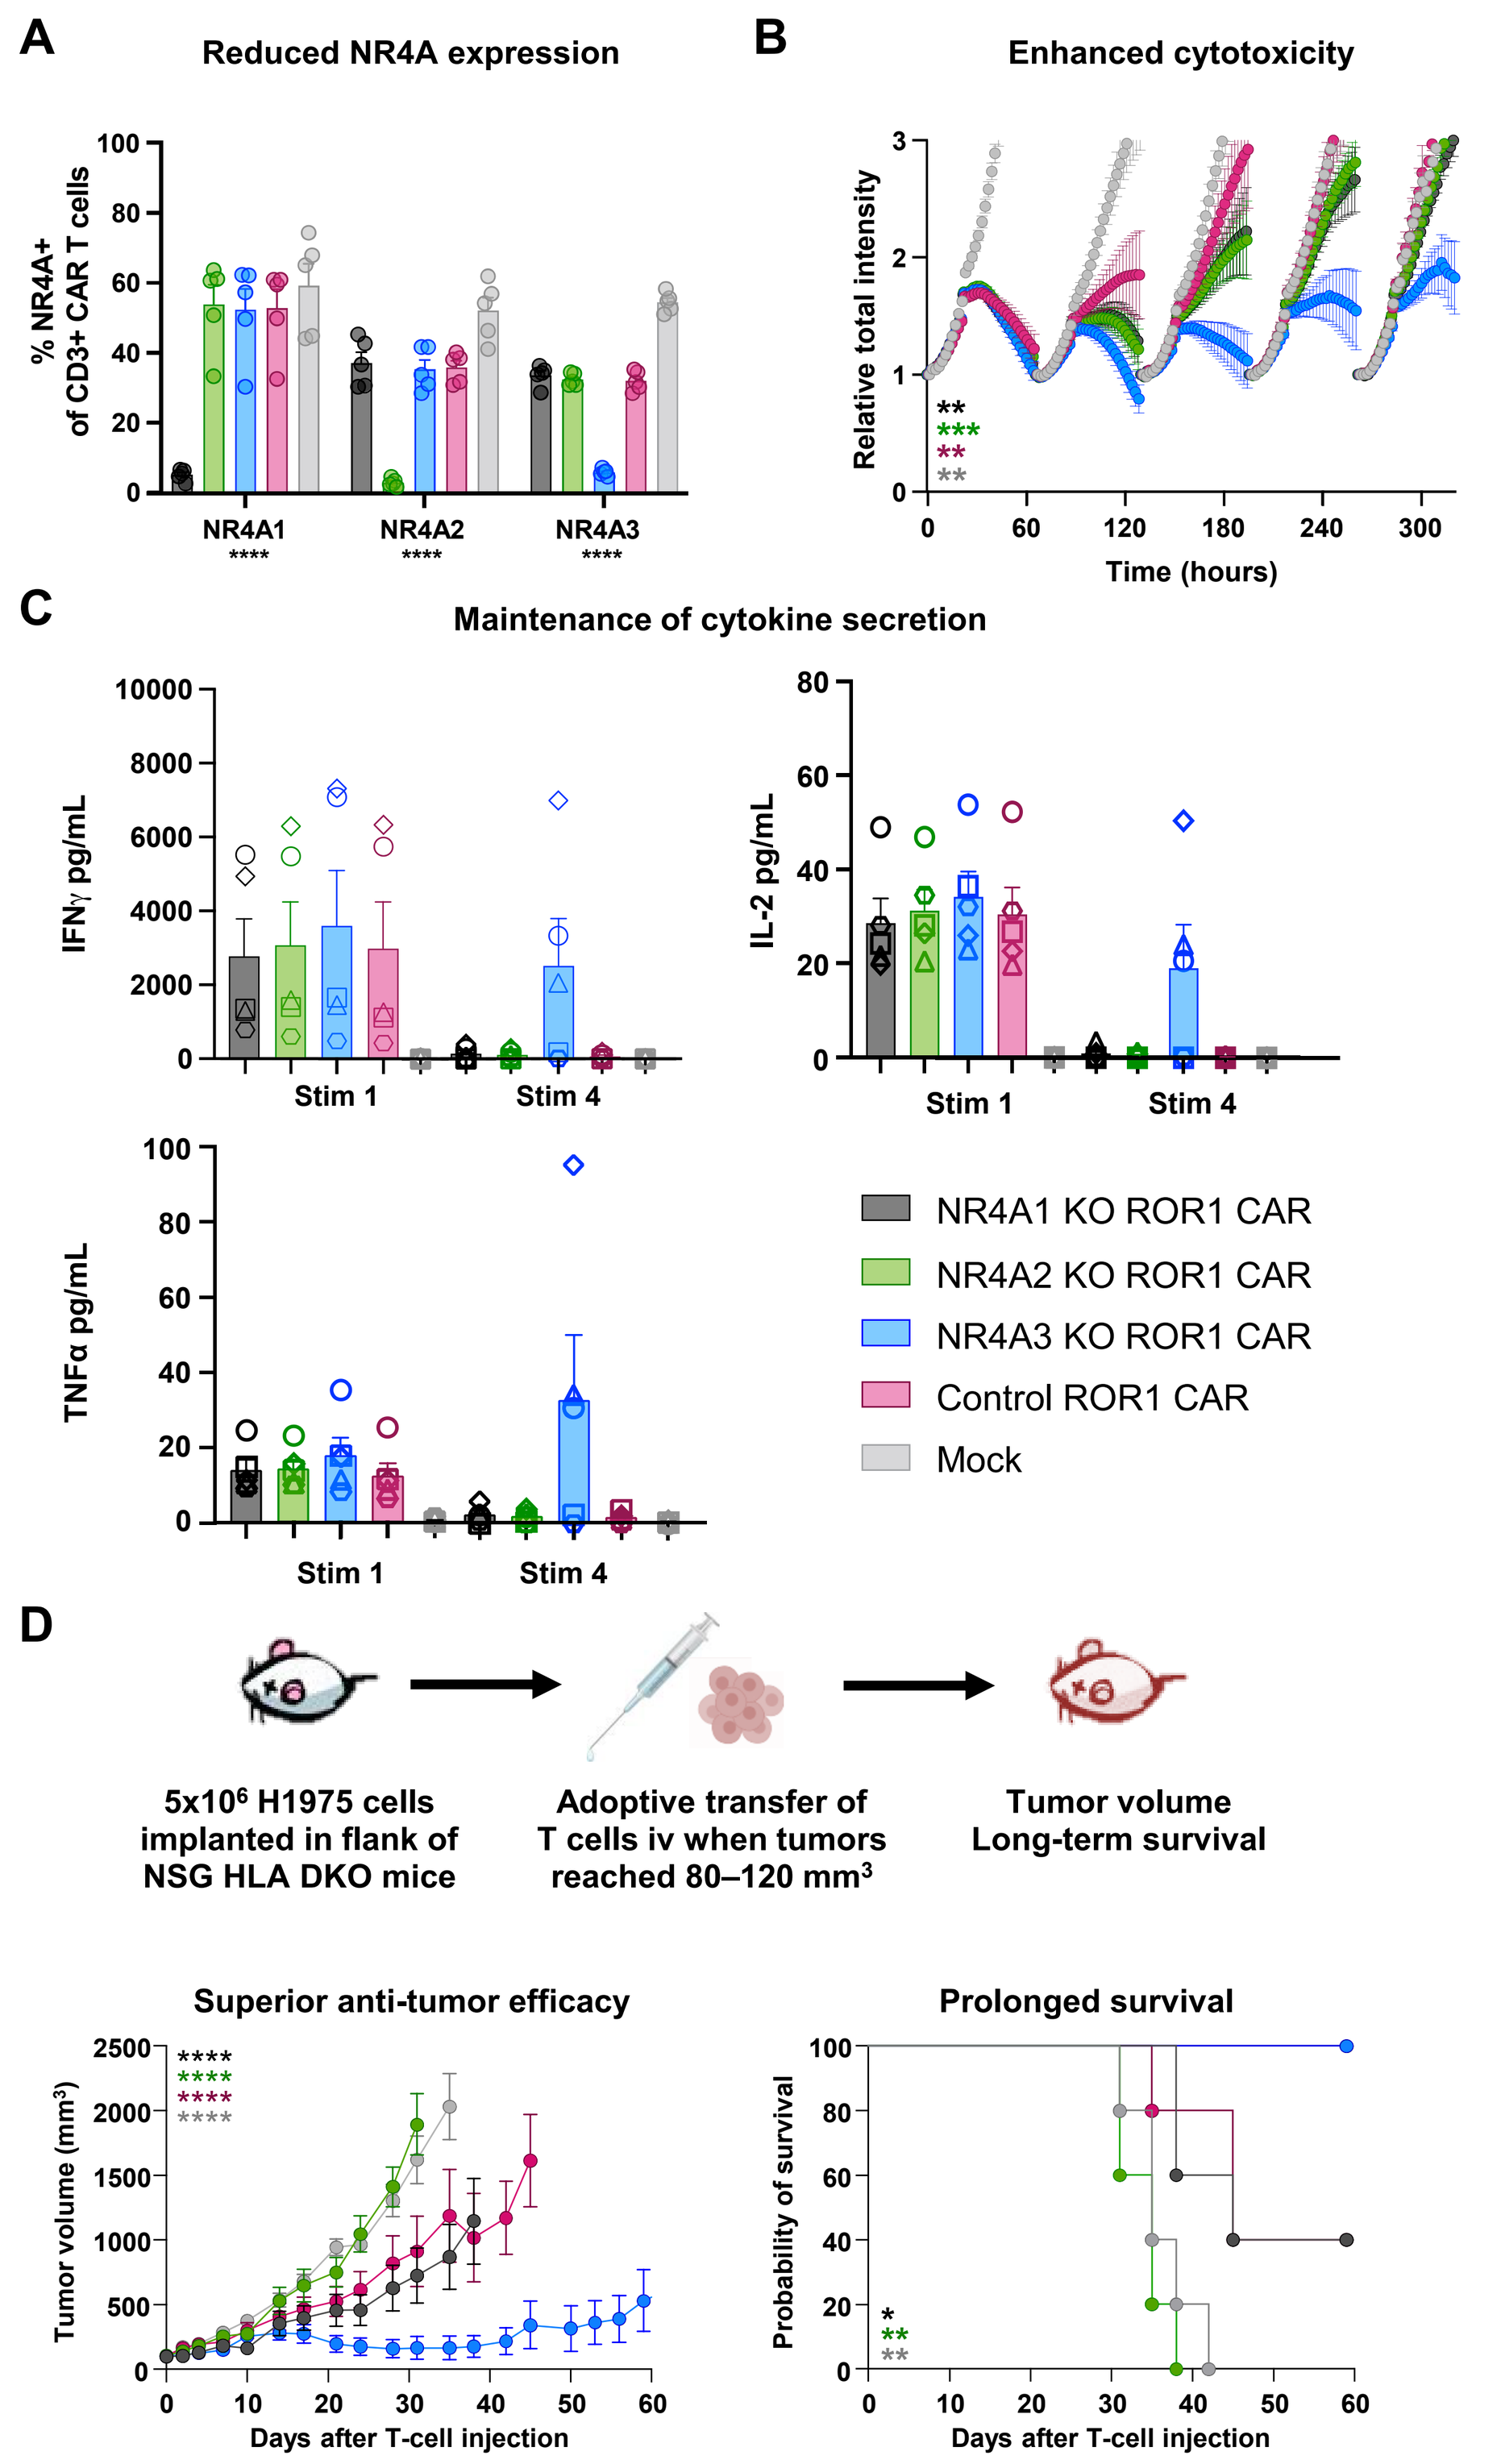
A
Reduced NR4A expression
% NR4A+ of CD3+ CAR T cells
NR4A1
NR4A2
NR4A3
B
Enhanced cytotoxicity
Relative total intensity
Time (hours)
C
Maintenance of cytokine secretion
IFNγ pg/mL
IL-2 pg/mL
TNFα pg/mL
Stim 1
Stim 4
NR4A1 KO ROR1 CAR
NR4A2 KO ROR1 CAR
NR4A3 KO ROR1 CAR
Control ROR1 CAR
Mock
D
5x10⁶ H1975 cells implanted in flank of NSG HLA DKO mice
Adoptive transfer of T cells iv when tumors reached 80–120 mm³
Tumor volume Long-term survival
Superior anti-tumor efficacy
Tumor volume (mm³)
Days after T-cell injection
Prolonged survival
Probability of survival

(A) NR4A protein expression was specifically reduced in *NR4A1* KO, *NR4A2* KO, and *NR4A3* KO CD3+ ROR1 CAR T cells compared to control unedited ROR1 CAR T cells and mock untransduced T cells on day 7 of production in five independent donors. Asterisks indicate significant differences between KO and unedited control. **(B)** Sequential stimulation with ROR1-expressing H1975-NucLightRed (NLR) target cells. Lysis of H1975-NLR target cells was quantified by measuring total NLR intensity. NLR intensity was normalized relative to the starting intensity after replating for each round of stimulation. Data were averaged for CAR T cells derived from 5 independent donors tested. Asterisks indicate significant differences at the last timepoint of the fifth stimulation compared to *NR4A3* KO. **(C)** Interferon gamma, Interleukin 2, and Tumor Necrosis Factor alpha (IFN-γ, IL-2, and TNF-α) secretion at Stim 1 and Stim 4 during the H1975 sequential stimulation assay. Open shapes represent the mean of triplicate wells for each donor of 5 independent donors tested. **(D)** Schematic of H1975 xenograft tumor model. Anti-tumor efficacy and survival associated with CAR T cells derived from 1 representative donor of 2 donors tested (n=5 mice/group). Tumor volume curves were truncated after removal of >20% of mice/group. Asterisks indicate significant differences compared to *NR4A3* KO. Error bars represent mean ± standard error of mean (SEM). **P<0.005; ***P<0.001; ****P<0.0001 by unpaired *t*-tests (A, B, C), Tukey's one-way ANOVA (D, left), or log-rank Manel-Cox test (D, right).

NR4A3 KO and c-Jun overexpression synergize to further reduce exhaustion and maintain ROR1 CAR T-cell function

- We generated *NR4A3* KO ROR1 CAR T cells with c-Jun overexpression (JUN–2A–ROR1 scFv-BBz–2A–EGFRt tri-cistronic vector) or without (CD19t–2A–ROR1 scFv-BBz–2A–EGFRt tri-cistronic vector) (**Figures 4 and 5**)
 - Control ROR1 CAR T-cells were edited at a control locus (human *CD19*) that is not expressed in T cells
 - All cells were generated using Lyell's epigenetic reprogramming technology (Epi-R)
 - *NR4A3* KO + c-Jun ROR1 CAR T-cell products were phenotypically indistinguishable from controls before antigen stimulation (data not shown) and were functionally comparable upon primary stimulation
 - *NR4A3* KO + c-Jun ROR1 CAR T-cell products exhibited improved tumor cell killing, high levels of cytokine production, increased CAR T-cell persistence, and reduced surface expression of inhibitory receptors after repetitive antigen stimulation, consistent with greater resistance to exhaustion-induced dysfunction (**Figure 6**)
- Transcriptomic analysis indicated that combination of *NR4A3* KO with c-Jun overexpression reduced the frequency of terminally exhausted ROR1 CAR T cells compared to control + c-Jun ROR1 CAR T cells following antigen restimulation (**Figure 7**)
- *NR4A3* KO + c-Jun ROR1 CAR T cells exhibited more robust anti-tumor efficacy *in vivo* (**Figure 8**)
 - Activity was observed with a 7-fold reduction in CAR T-cell dose compared to controls
 - *NR4A3* KO + c-Jun ROR1 CAR T cells demonstrated a 10-fold greater expansion in blood compared to control + c-Jun ROR1 CAR T cells

Figure 4: Schematic of *NR4A3* KO + c-Jun ROR1 CAR T cells

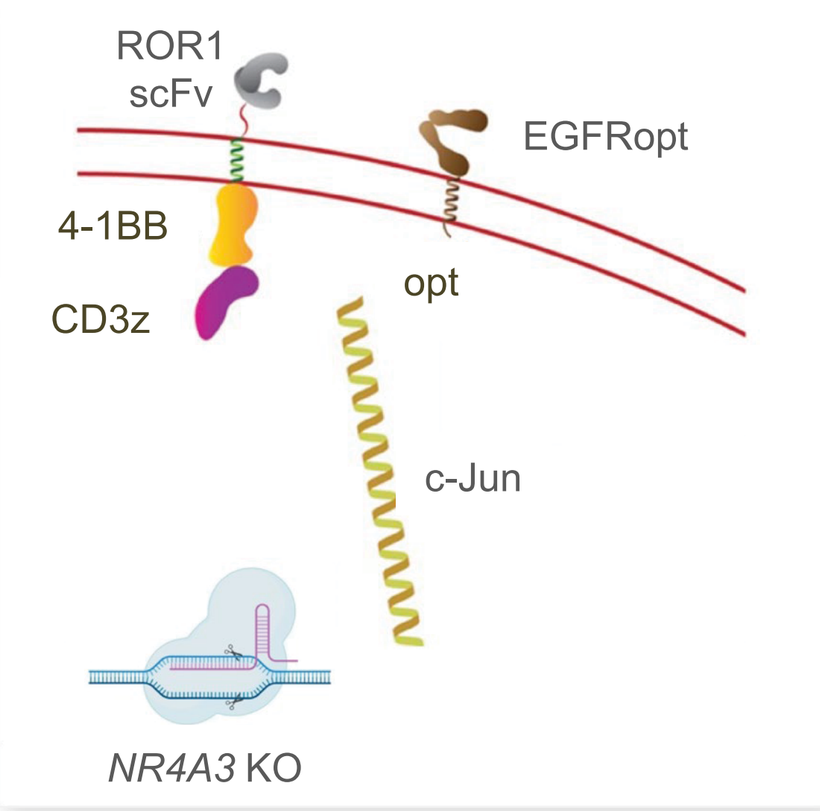
ROR1 scFv
4-1BB
CD3z
EGFRopt
opt
c-Jun
NR4A3 KO

Figure 5: Efficient *NR4A3* editing in c-Jun–overexpressing ROR1 CAR T cells

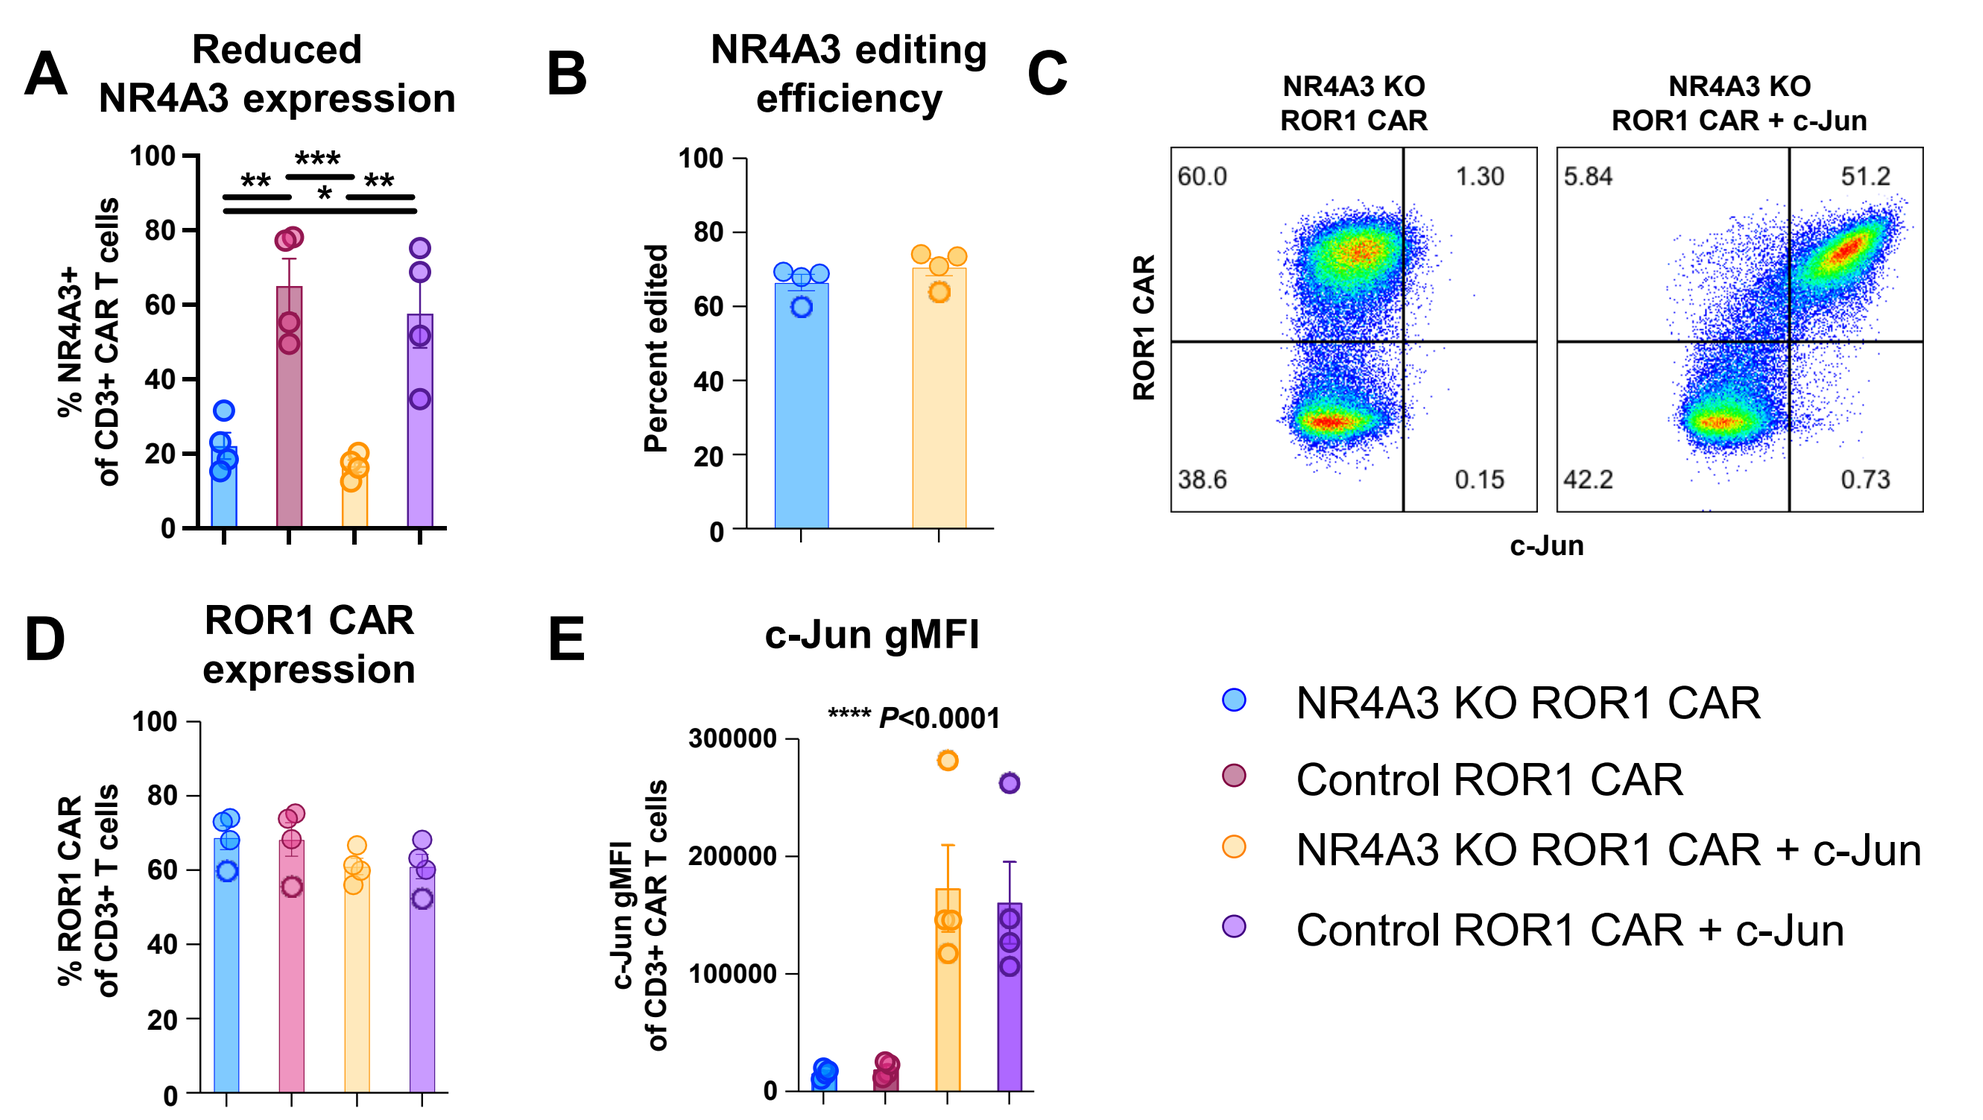
A
Reduced NR4A3 expression
% NR4A3+ of CD3+ CAR T cells
B
NR4A3 editing efficiency
Percent edited
C
NR4A3 KO ROR1 CAR
NR4A3 KO ROR1 CAR + c-Jun
ROR1 CAR
c-Jun
D
ROR1 CAR expression
% ROR1 CAR of CD3+ T cells
E
c-Jun gMFI
**** P<0.0001
c-Jun gMFI of CD3+ CAR T cells
NR4A3 KO ROR1 CAR
Control ROR1 CAR
NR4A3 KO ROR1 CAR + c-Jun
Control ROR1 CAR + c-Jun

(A) NR4A3 protein expression was significantly reduced by 60–70% in control or c-Jun overexpressing ROR1 CAR T cells derived from 4 independent donors tested. **(B)** *NR4A3* genomic editing efficiency based on NGS was reflective of NR4A3 protein expression. **(C)** Representative flow cytometry plot showing ROR1 CAR and c-Jun co-expression in CD3+ T cells derived from a single donor. *NR4A3* editing does not affect expression of ROR1 CAR **(D)** or c-Jun **(E)**. Asterisks indicate significant differences comparing ROR1 CAR T cells with and without c-Jun overexpression independent of *NR4A3* editing. Error bars represent mean ± SEM.

*P<0.05; **P<0.005; ***P<0.001; ****P<0.0001 by unpaired *t*-test

Figure 6: *NR4A3* KO and c-Jun overexpression synergize to enhance ROR1 CAR T-cell function *in vitro*

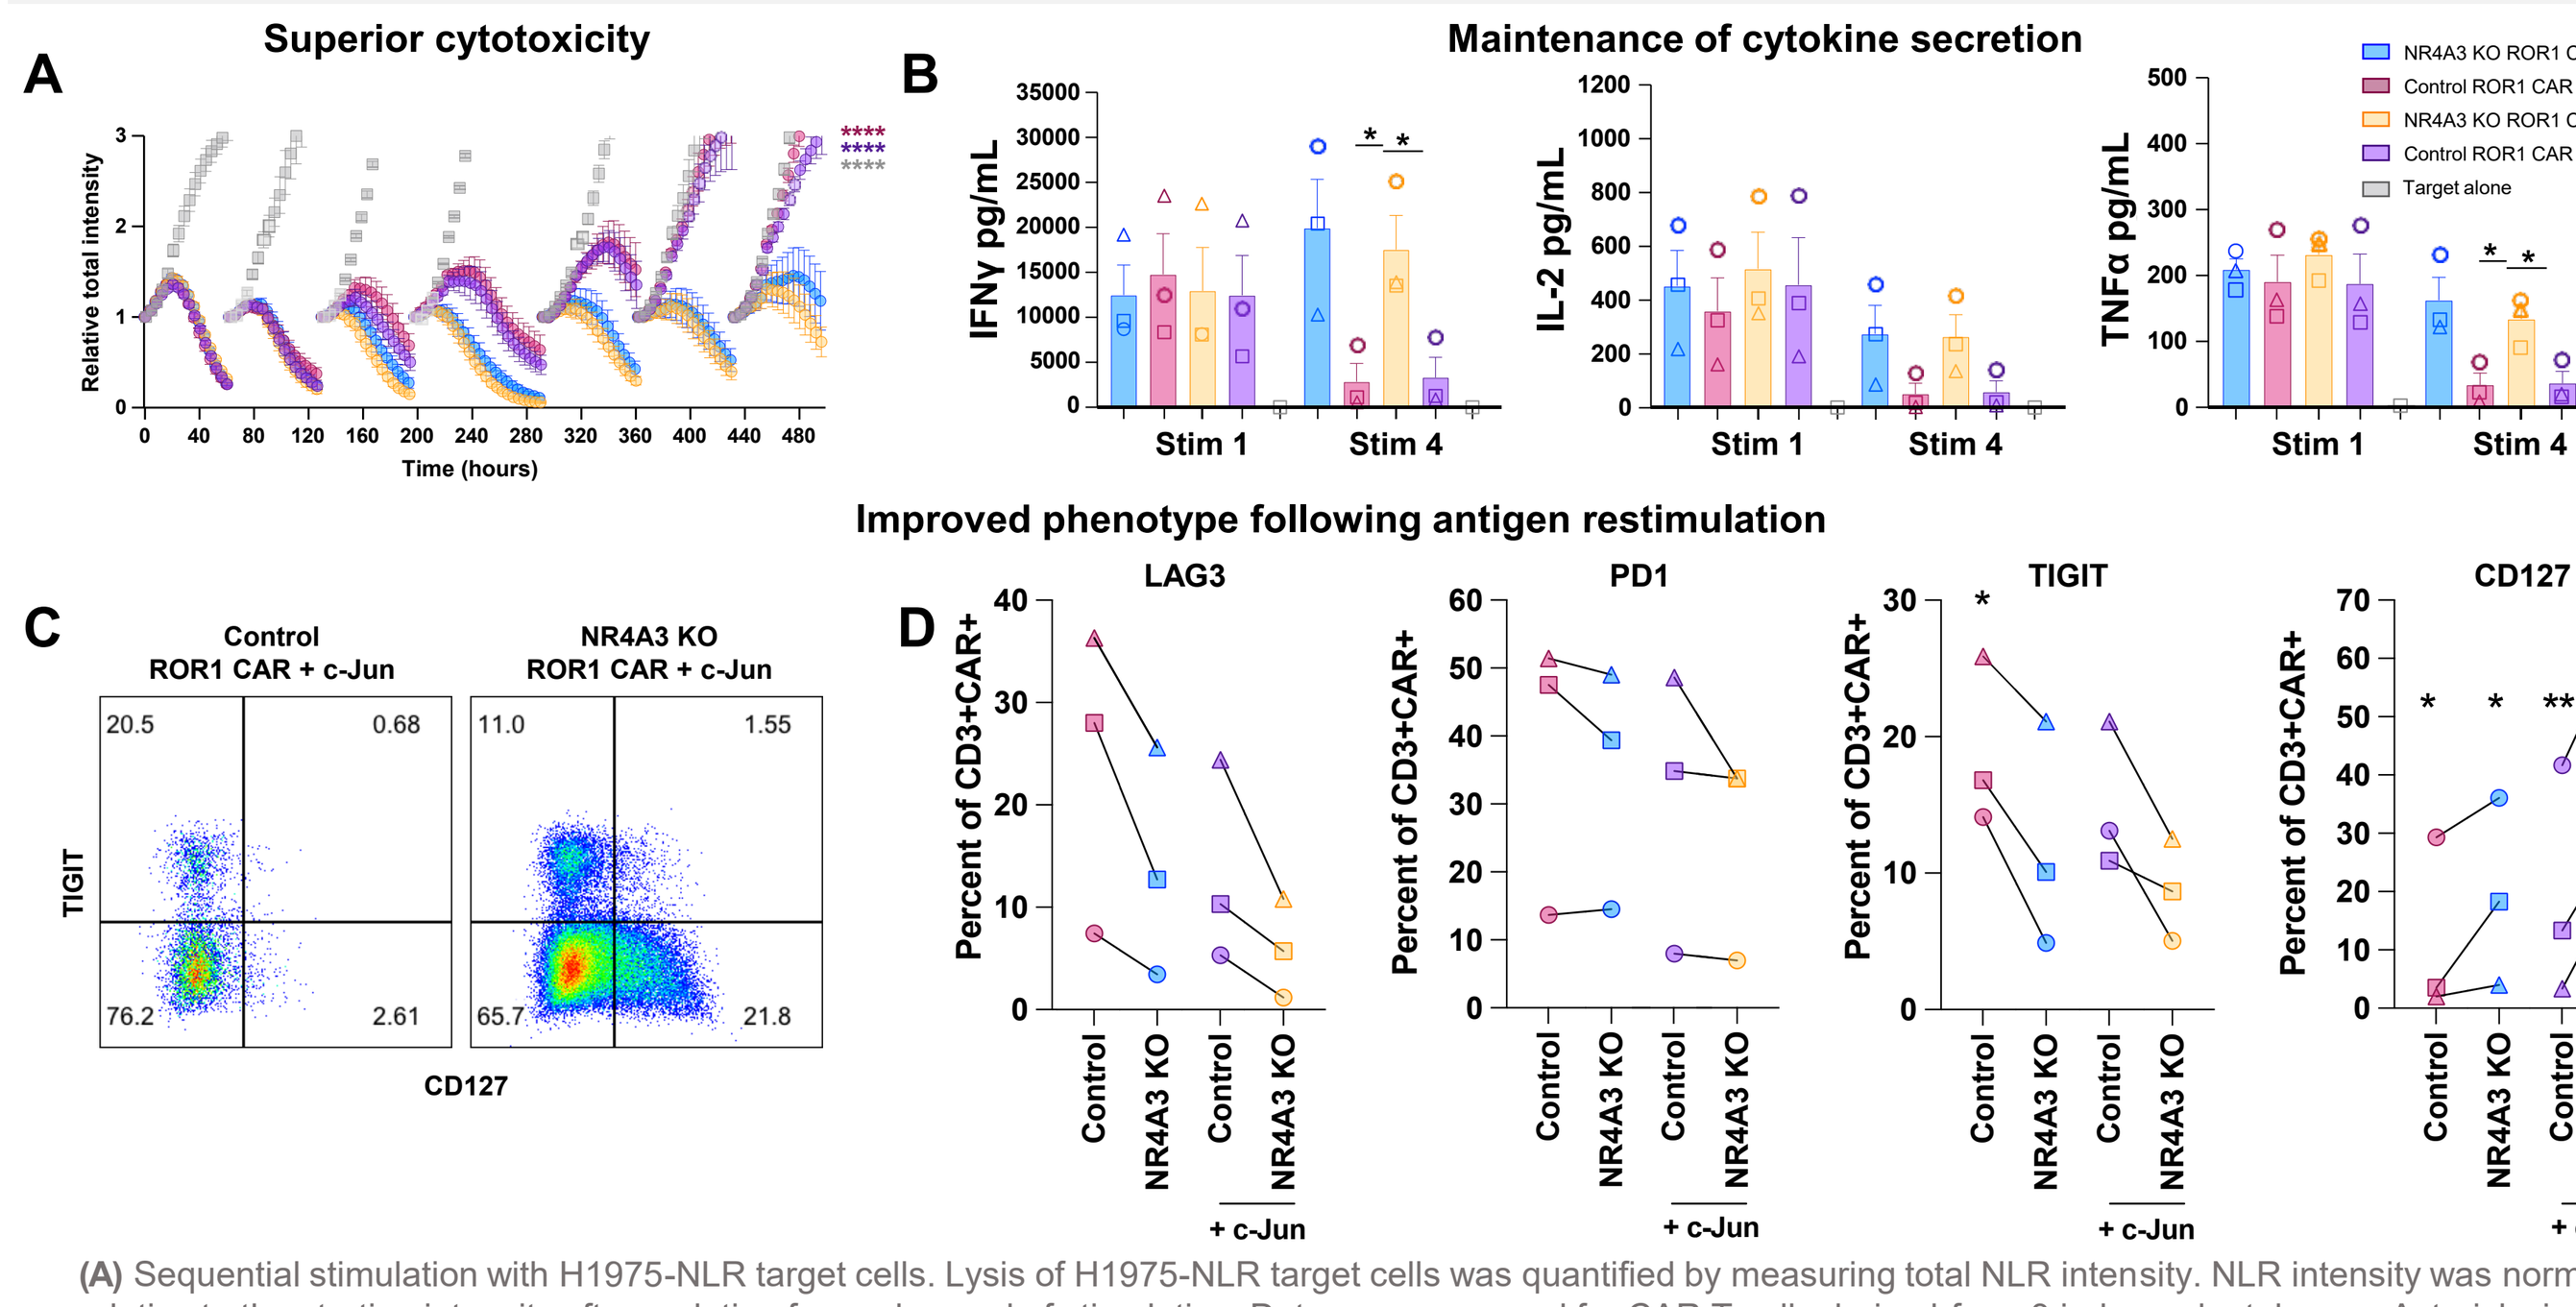
A
Superior cytotoxicity
Relative total intensity
Time (hours)
B
Maintenance of cytokine secretion
IFNγ pg/mL
IL-2 pg/mL
TNFα pg/mL
Stim 1
Stim 4
NR4A3 KO ROR1 CAR
Control ROR1 CAR
NR4A3 KO ROR1 CAR + c-Jun
Control ROR1 CAR + c-Jun
Target alone
Improved phenotype following antigen restimulation
C
Control ROR1 CAR + c-Jun
NR4A3 KO ROR1 CAR + c-Jun
TIGIT
CD127
D
LAG3
PD1
TIGIT
CD127
Percent of CD3+CAR+
Control
NR4A3 KO
+ c-Jun

(A) Sequential stimulation with H1975-NLR target cells. Lysis of H1975-NLR target cells was quantified by measuring total NLR intensity. NLR intensity was normalized relative to the starting intensity after replating for each round of stimulation. Data were averaged for CAR T cells derived from 3 independent donors. Asterisks indicate significant differences compared to *NR4A3* KO + c-Jun at the last timepoint of the seventh stimulation. **(B)** IFN-γ, IL-2, and TNF-α secretion at Stim 1 and Stim 4 during the H1975 sequential stimulation assay. Open shapes represent the mean of triplicate wells for each of 3 independent donors tested. Asterisks indicate significant differences compared to *NR4A3* KO + c-Jun ROR1 CAR T cells at the fourth stimulation. **(C)** Representative flow cytometry plot showing TIGIT and CD127 expression of CD3+ROR1 CAR+ T cells derived from a single donor corresponding to the fourth stimulation in (A). **(D)** Surface expression of inhibitory receptors and CD127 (IL7R) after the fourth stimulation in (A). Each shape corresponds to CAR T cells derived from one of 3 independent donors tested. Asterisks indicate significant differences compared to *NR4A3* KO + c-Jun ROR1 CAR T cells. Error bars represent mean ± SEM. *P<0.05; **P<0.005; ****P<0.0001 by unpaired *t*-test (A, B) or paired *t*-test (D).

Figure 7: Transcriptomic analysis of *NR4A3* KO + c-Jun CD8+ ROR1 CAR T cells revealed reduced terminal exhaustion and enhanced memory-like cell proportions following serial restimulation with A549

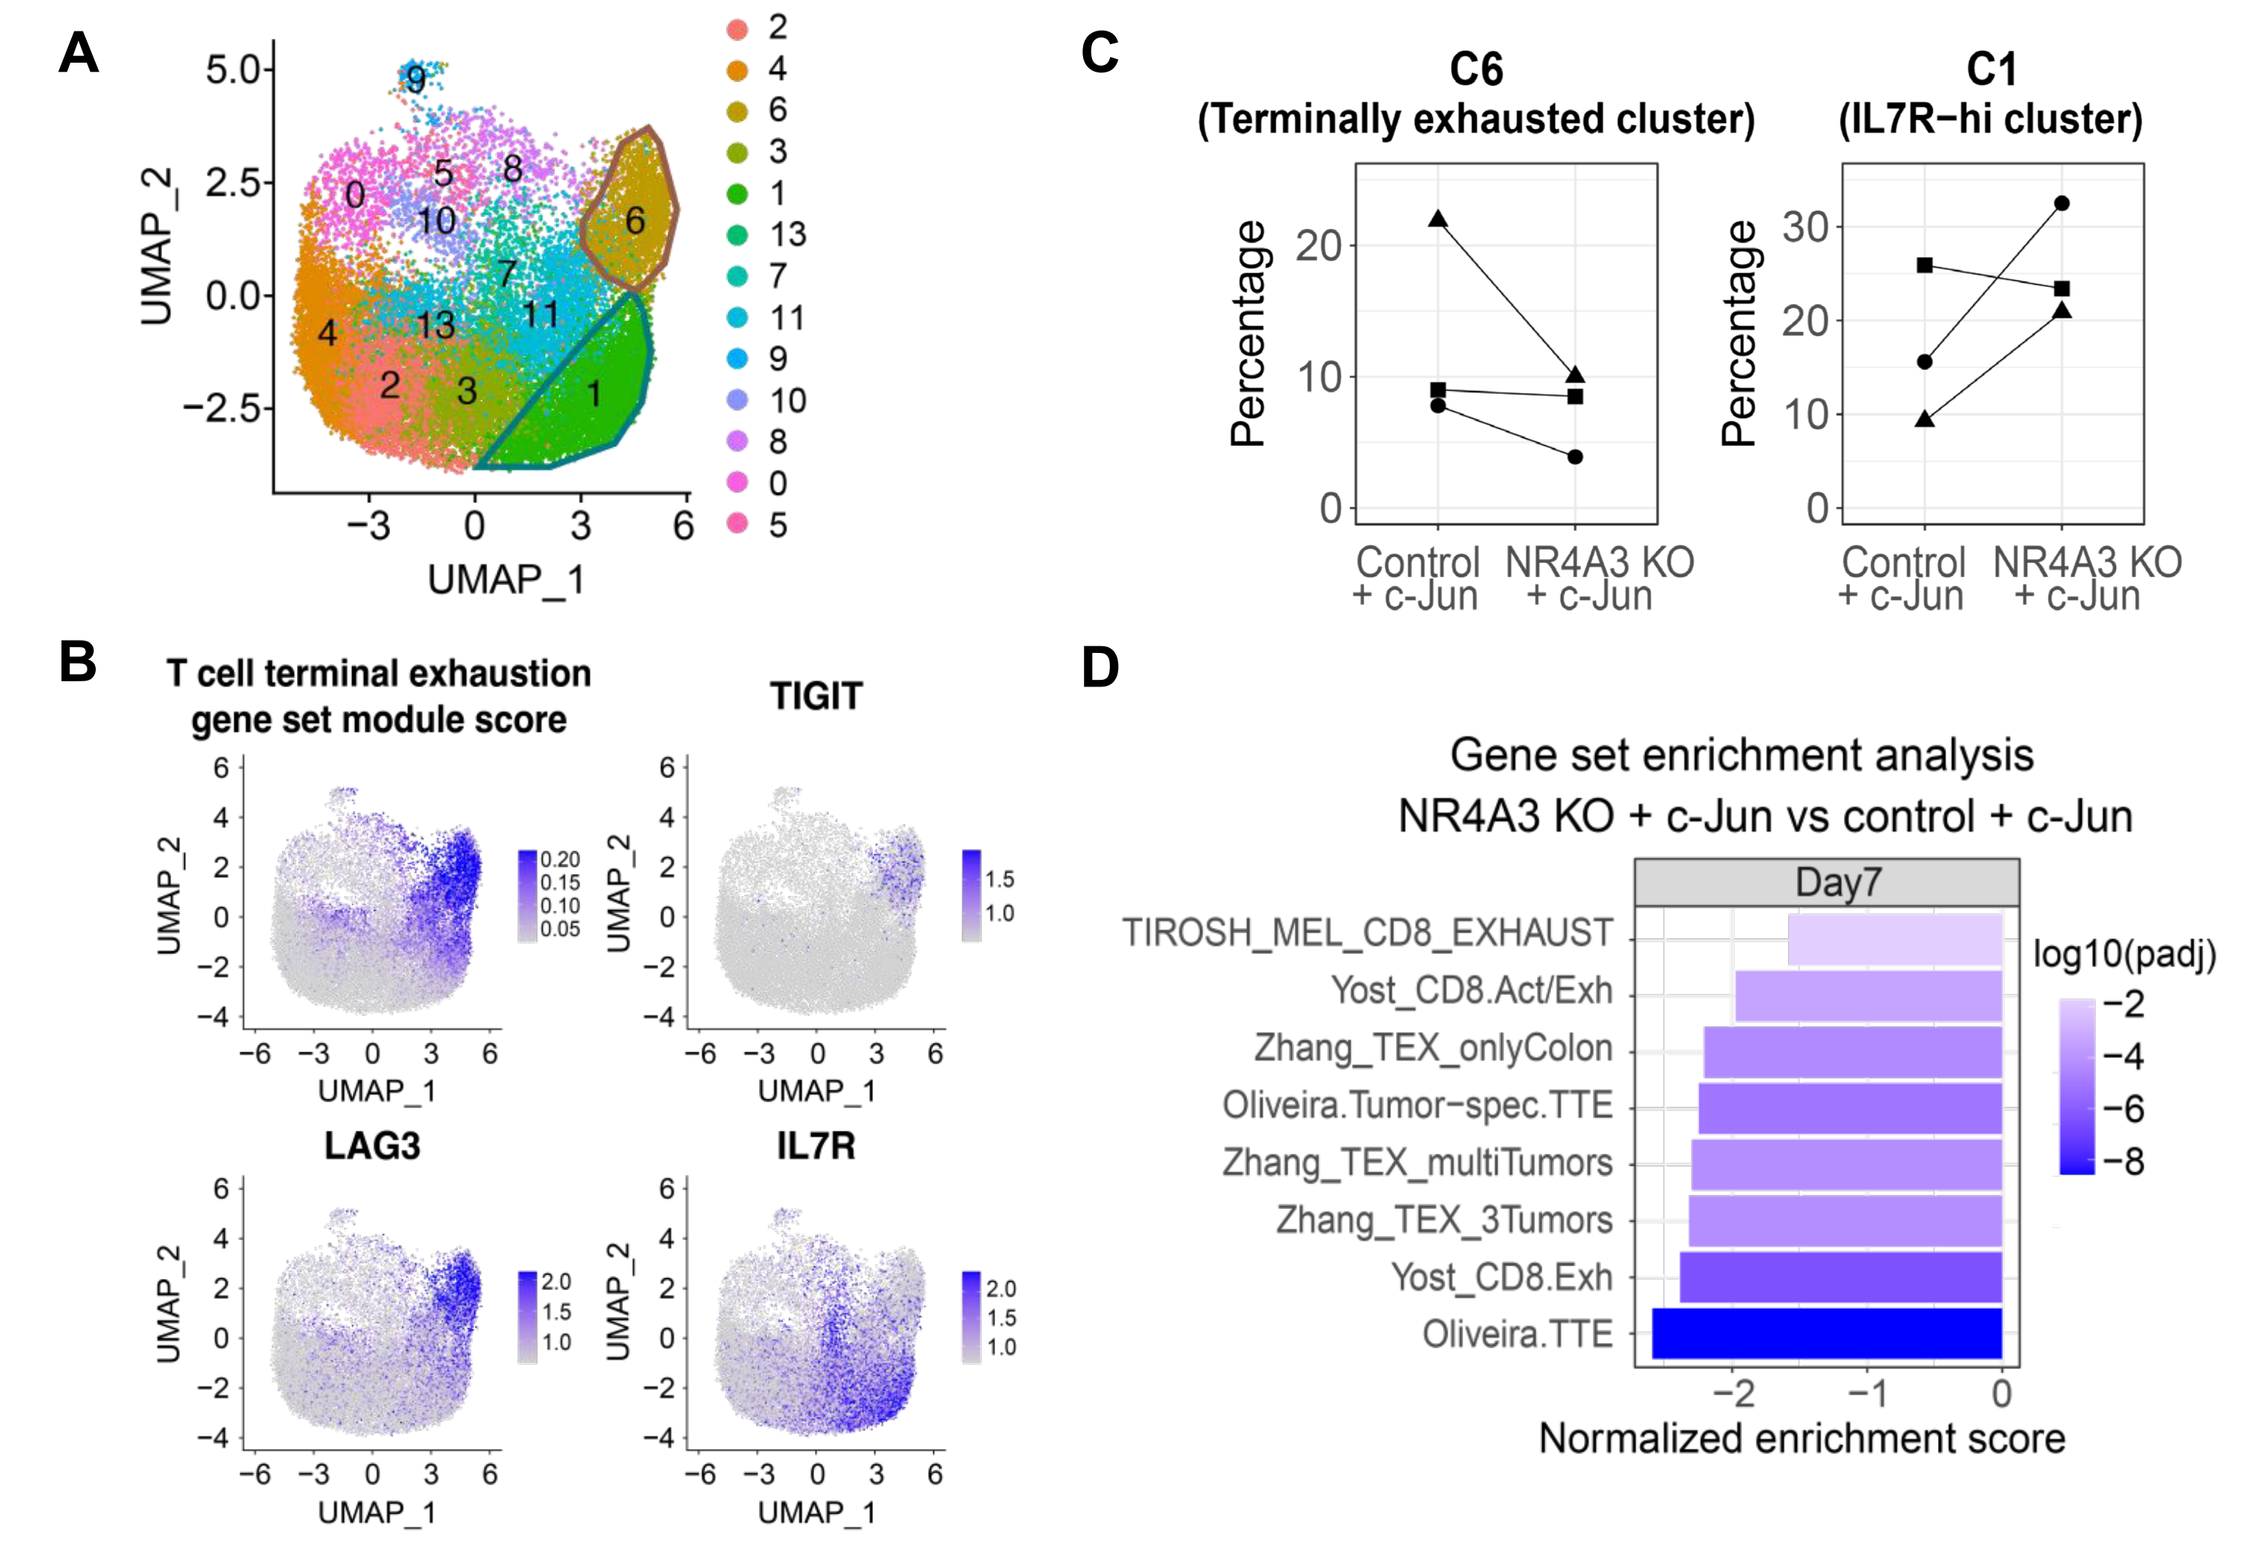
A
UMAP_1
UMAP_2
B
T cell terminal exhaustion gene set module score
TIGIT
LAG3
IL7R
C
C6 (Terminally exhausted cluster)
C1 (IL7R−hi cluster)
Percentage
Control + c-Jun
NR4A3 KO + c-Jun
D
Gene set enrichment analysis
NR4A3 KO + c-Jun vs control + c-Jun
Day7
TIROSH_MEL_CD8_EXHAUST
Yost_CD8.Act/Exh
Zhang_TEX_onlyColon
Oliveira.Tumor−spec.TTE
Zhang_TEX_multiTumors
Zhang_TEX_3Tumors
Yost_CD8.Exh
Oliveira.TTE
log10(padj)
Normalized enrichment score

Single-cell and bulk RNA-Seq of *NR4A3* KO ROR1 CAR T cells and control-edited CD8+ ROR1 CAR T cells with c-Jun overexpression following 7 days of *in vitro* antigen restimulation. A549 target cells were used for restimulation, as this model has been previously shown to rapidly induce features of exhaustion *in vitro*. **(A)** Single cell RNA-Seq clustering analysis identified the terminally exhausted cluster (C6) highlighted in brown and the IL7R-hi cluster (C1) highlighted in green. **(B)** Cluster C6 is enriched for module score of a previously published T-cell terminal exhaustion gene set[7] and for RNA expressions of TIGIT and LAG3, whereas cluster C1 is enriched for expression of IL7R. **(C)** The proportion of cells in the terminally exhausted cluster (C6) was reduced, and the proportion in the IL7R-hi cluster (C1) was increased in *NR4A3* KO + c-Jun CD8+ ROR1 CAR T cells compared to control-edited + c-Jun CD8+ ROR1 CAR T cells. Closed shapes represent CAR T cells derived from each of 3 independent donors tested. **(D)** *NR4A3* KO + c-Jun CD8+ ROR1 CAR T cells exhibited depletion of exhaustion-associated gene sets compared to control + c-Jun ROR1 CAR T cells based on bulk RNA-Seq data. Data represent CAR T cells derived from 3 independent donors tested.

Figure 8: *NR4A3* KO + c-Jun ROR1 CAR T cells exhibited robust anti-tumor activity *in vivo*

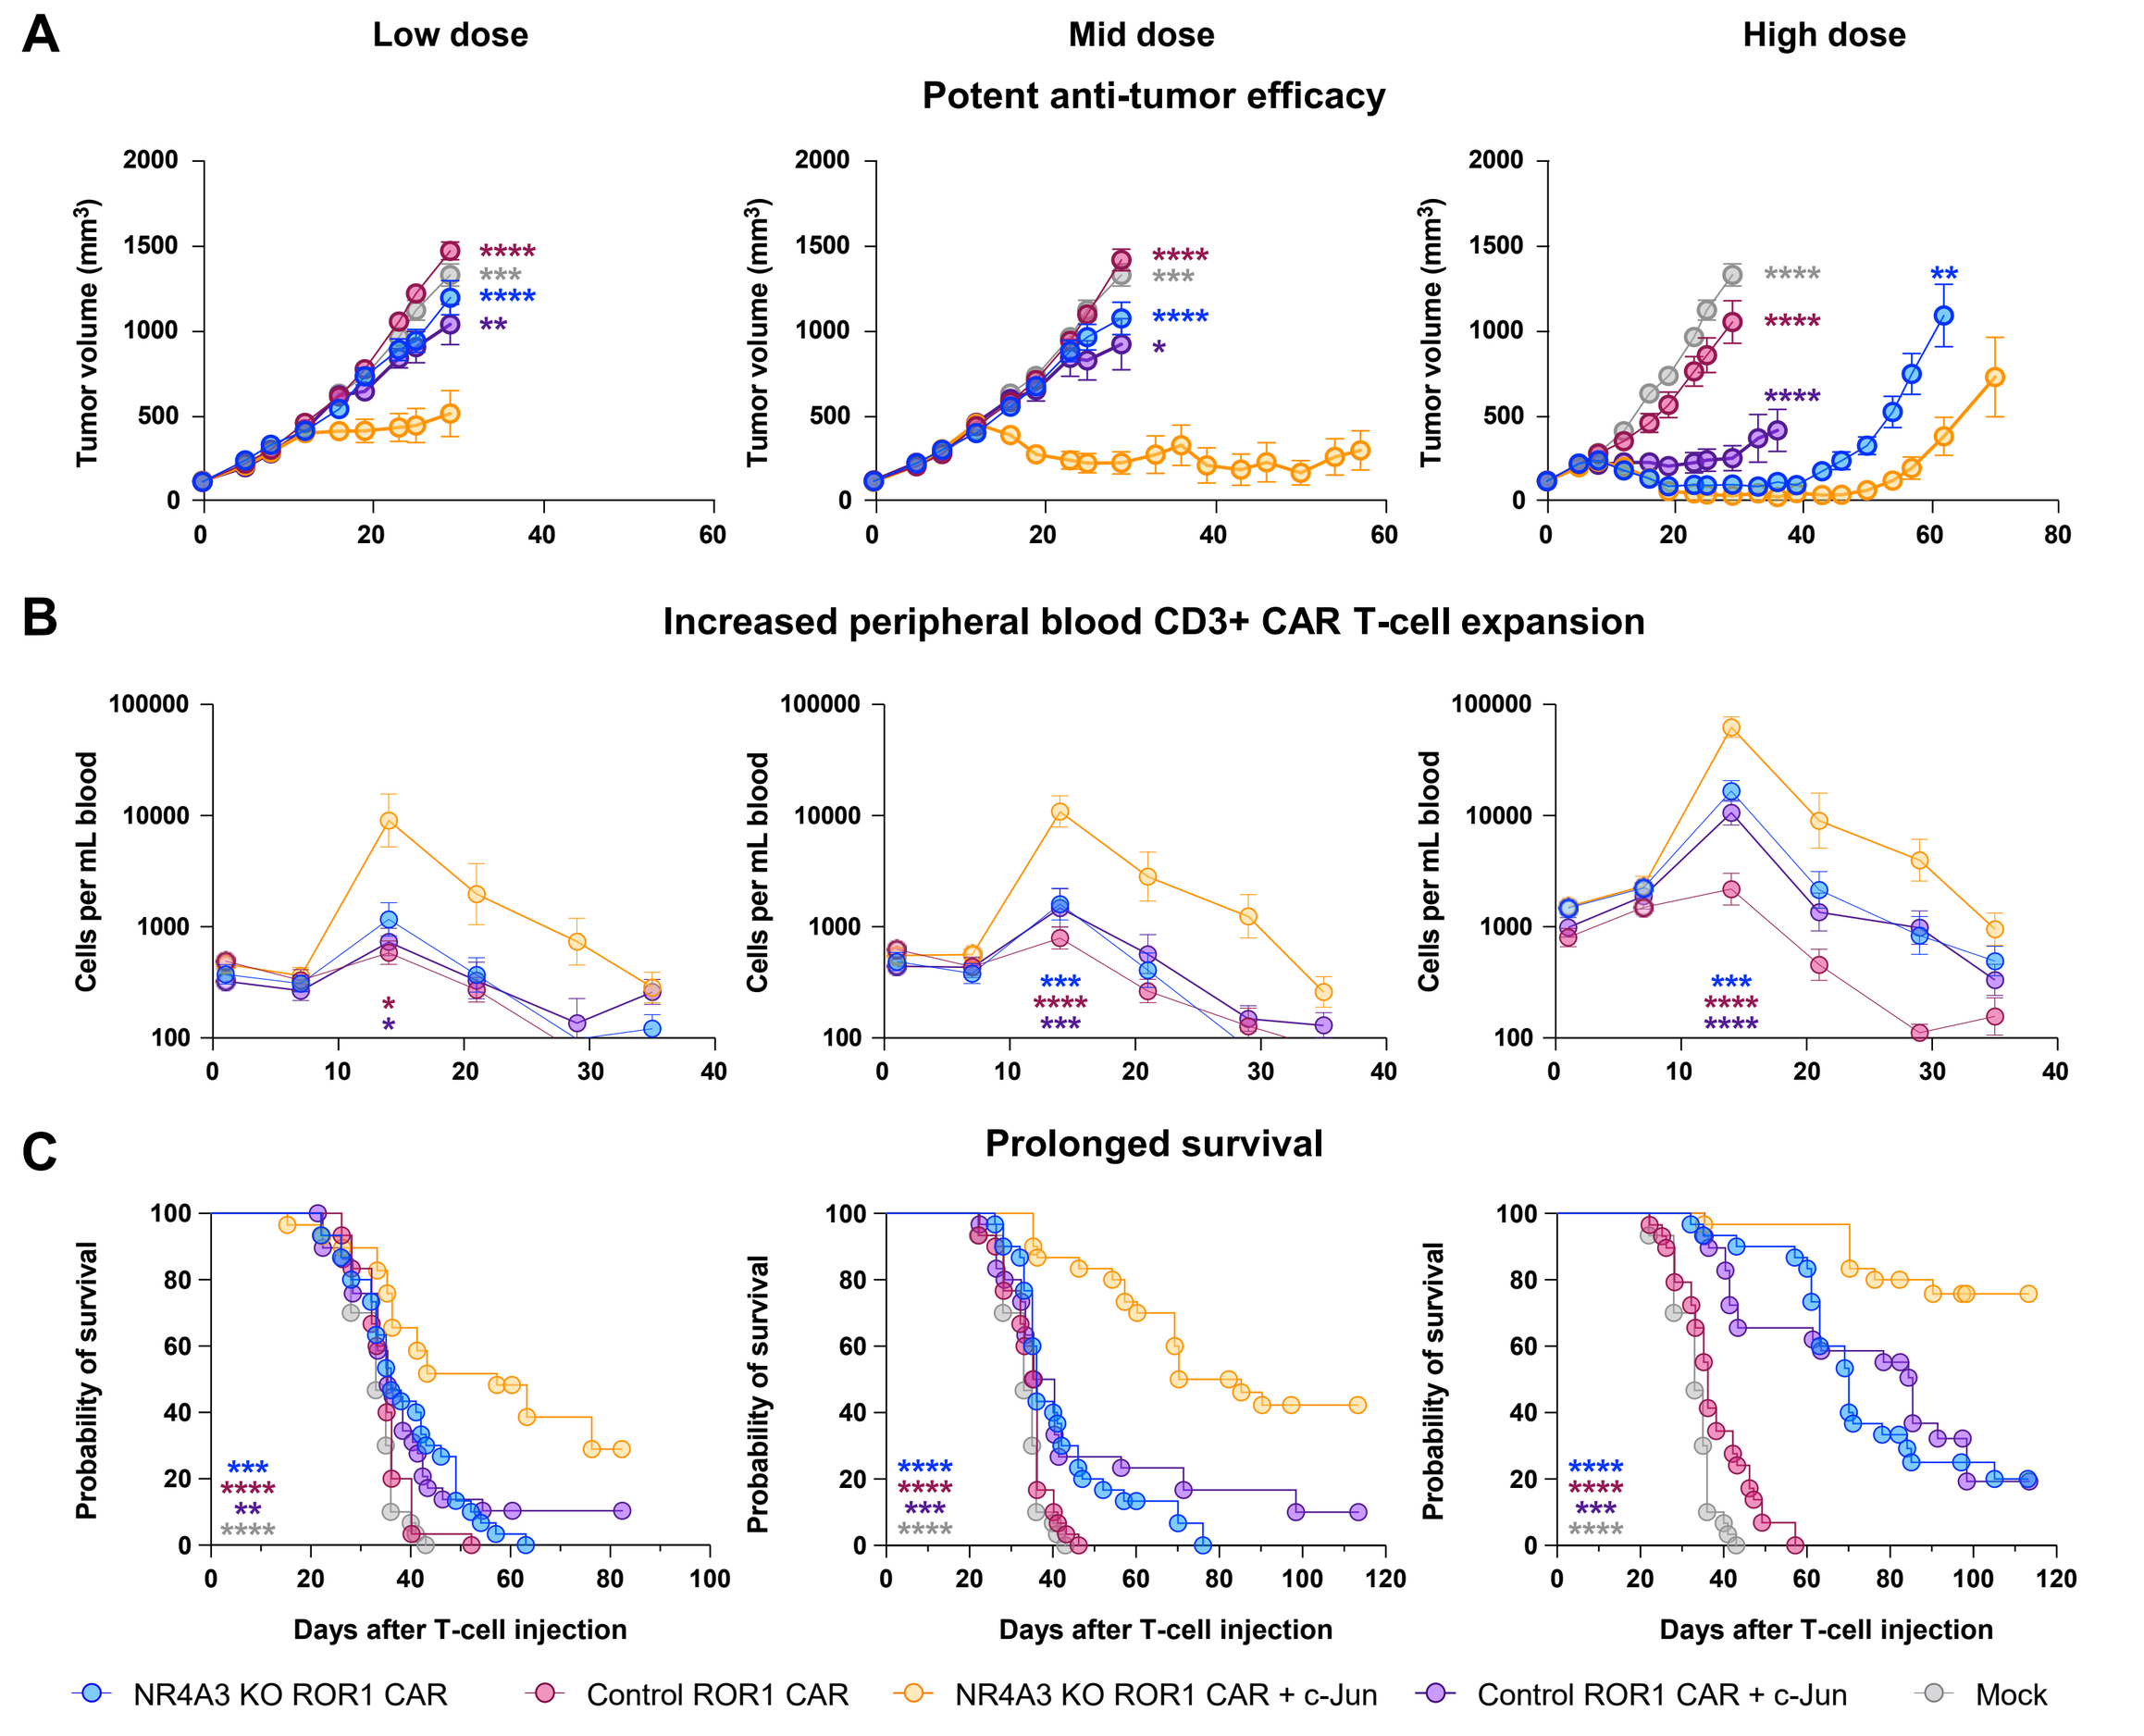
A
Low dose
Mid dose
High dose
Potent anti-tumor efficacy
Tumor volume (mm³)
B
Increased peripheral blood CD3+ CAR T-cell expansion
Cells per mL blood
C
Prolonged survival
Probability of survival
Days after T-cell injection
NR4A3 KO ROR1 CAR
Control ROR1 CAR
NR4A3 KO ROR1 CAR + c-Jun
Control ROR1 CAR + c-Jun
Mock

(A) Average tumor volumes for NSG HLA dKO mice implanted with subcutaneous flank H1975 xenograft tumors. Mice were subjected to intravenous (IV) injection with 2x10⁶ mock untransduced unedited T cells, *NR4A3* KO ROR1 CAR T cells, or control-edited ROR1 CAR T cells with or without c-Jun overexpression at either 0.3x10⁶ (left, low dose), 0.6x10⁶ (middle, mid dose), or 2x10⁶ (right, high dose) CAR T cells per mouse when mean tumor volumes reached 80-120 mm³. Tumor volume curves were truncated after removal of >20% of mice/group. Asterisks indicate significant differences compared to *NR4A3* KO + c-Jun ROR1 CAR T-cell–treated animals. **(B)** Average numbers of peripheral blood CD3+CAR+ T cells derived from animals treated with CAR T cells. Asterisks indicate significant differences at day 14 post T-cell injection compared to *NR4A3* KO + c-Jun ROR1 CAR T-cell–treated animals. **(C)** Survival of tumor-bearing mice at all CAR T-cell doses tested. Asterisks indicate significant differences compared to *NR4A3* KO + c-Jun ROR1 CAR T-cell–treated animals. Data were averaged for CAR T-cells derived from 3 independent donors tested with n=10 mice/group for each donor. Error bars represent mean ± SEM. *P<0.05; **P<0.005; ***P<0.001; ****P<0.0001 by Tukey's one-way ANOVA (A), unpaired *t*-test (B), or log-rank Mantel-Cox test (C). (Data not shown) Consistent with *in vitro* transcriptomic analysis results, *NR4A3* KO + c-Jun ROR1 CAR T cells exhibited reduced proportions of terminally exhausted cells and increased proportions of IL7R-hi memory-like cells in the *in vivo* H1975 xenograft model on day 14 post T-cell injection compared to control-edited + c-Jun ROR1 CAR T cells (n=2 mice/group, 1 donor, 2.5x10⁶ CAR T-cell dose).

Conclusions

- **ROR1 CAR T cells with *NR4A3* KO maintain functional activity following repeat antigen stimulation *in vitro* and *in vivo* compared to control unedited, *NR4A1* KO, or *NR4A2* KO ROR1 CAR T cells**
- ***NR4A3* KO + c-Jun ROR1 CAR T cells exhibited:**
 - **Enhanced and sustained killing activity compared to control + c-Jun ROR1 CAR T cells**
 - **Higher levels of cytokines after later rounds of stimulation compared to control + c-Jun ROR1 CAR T cells**
 - **Reduced terminally exhausted cell population and increased IL7R+ cell population compared to control + c-Jun ROR1 CAR T cells**
 - **Enhanced anti-tumor potency with tumor control at a ~7-fold lower CAR T-cell dose**
 - **Enhanced *in vivo* expansion with >10-fold higher peak numbers of CAR T cells, which contracted upon tumor clearance**
- **Combining reduction of *NR4A3* expression with overexpression of c-Jun has the potential to further limit T-cell exhaustion and provide durable ROR1 CAR T-cell functional activity compared to either strategy alone**
- **To test whether this strategy can enhance the efficacy of cellular immunotherapies targeting ROR1-expressing solid tumors, Lyell has initiated preclinical development of LYL119, an investigational ROR1 targeting CAR-T cell therapy that incorporates both of these genetic reprogramming technologies**

References

1. Beatty GL, O'Hara M. *Pharmacol Ther*. 2016;166:30-39. 2. Fraietta JA, et al. *Nat Med*. 2018;24:563-571. 3. Lynn RC, et al. *Nature*. 2019;576:293-300. 4. Park S, et al. AACR Annual Meeting 2022 Poster. 5. Chen J, et al. *Nature*. 2019;567:530-534. 6. Tirosh I, et al. *Science*. 2016;352:189-196. 7. Liu X, et al. *Nature*. 2019;567:525-529. 8. Odagiu I, et al. *Proc Natl Acad Sci USA*. 2020;117:24392-24402.

Acknowledgments

We thank members of Lyell Immunopharma's *In Vivo* Pharmacology team (Shawn Anderson, Jacob Corpuz, and Quinn Walker), NGS/Flow team (Amir Figueroa, Elizabeth Hernandez, Andrew Jimena, Amanda Sims, Emily Fu-Sum, Yi-Dong Lin, Sheila Lou, Stefan Siebert, Ken Xiong, and Lora Zhao), Research team (Rachel Fukuda, Helle Jensen, Megan Murt, Spencer Park, Blythe Sather, Courtney Simianer, Sydney Spadinger, and Nicole Zhang), and Vector Sciences team (David Anderson, Martin Wohlfahrt, and Chang-Chi Wu) for their experimental contributions. Medical writing and editorial support were funded by Lyell Immunopharma, Inc and provided by Melanie Styers, PhD, of Verascity Science.

Ethics Statement

Experiments presented in this abstract relied on human donor material that was obtained from commercial vendors. These vendors use their own IRB-approved protocol and consent process. *In vivo* experiments presented in this abstract were approved by Lyell Immunopharma's IACUC (EB17-010-117).

Supplementary Materials

Please scan the Quick Response (QR) code to download poster. Copies of content obtained through the QR code are for personal use only and may not be reproduced without permission from the author of this poster: Viola Lam (vlam@lyell.com)

Presented at SITC Annual Meeting 2022; Nov 8–12; Boston, MA, USA